EOS884448, a high affinity fully human antibody directed against TIGIT, mediates in vitro anti-tumor activity through multiples mechanisms of action involving activation of intratumor effector cells and depletion of regulatory T cells.

Cuende J [1], Rabolli V [1], Nyawouame F [1], Mercier M [1], Pappalardo A [2], Garnero L [1], Dechanet-Merville J [2], Preillon J [1], Hoofd C [1] and Driessens G [1].

[1] iTeos Therapeutics
[2] Immunoconcept, University of Bordeaux, Fr

AACR 2019 #3240

SUMMARY

- T cell Immunoreceptor with Ig and ITIM domains (TIGIT) is a negative costimulatory receptor that inhibits effector T cell and NK cell function and marks a highly suppressive regulatory T cell (Treg) subset.
- TIGIT ligands belong to the PVR/nectin family, among which PVR (CD155) shows the highest affinity and is commonly expressed on antigen presenting cells (APC) and tumor cells. *See poster #4969.*
- CD226, a co-stimulatory receptor also expressed on NK and T cells, competes with TIGIT for PVR binding but with a lower affinity.
- TIGIT expression is increased on T and NK(T) cells from cancer patients and is correlated to poor outcome and response to aPD1 therapy in some indications.
- EOS884448 properties and functionality make it an attractive Immuno-Oncology therapy candidate:
 - ✓ Strong binding to primary human and cyno T cells (sub-nM Kd)
 - ✓ Competition with natural ligands with IC_{50} in sub-nM range
 - ✓ Increase of primary T cell functions in healthy donors and cancer patients
 - ✓ Depletion through ADCC of highly suppressive TIGIT⁺ Tregs.
 - ✓ Antitumor efficacy in animal model
 - ✓ Excellent developability profile
 - ✓ Excellent safety profile

EOS884448 PREVENTS TIGIT-DRIVEN IMMUNOSUPPRESSION

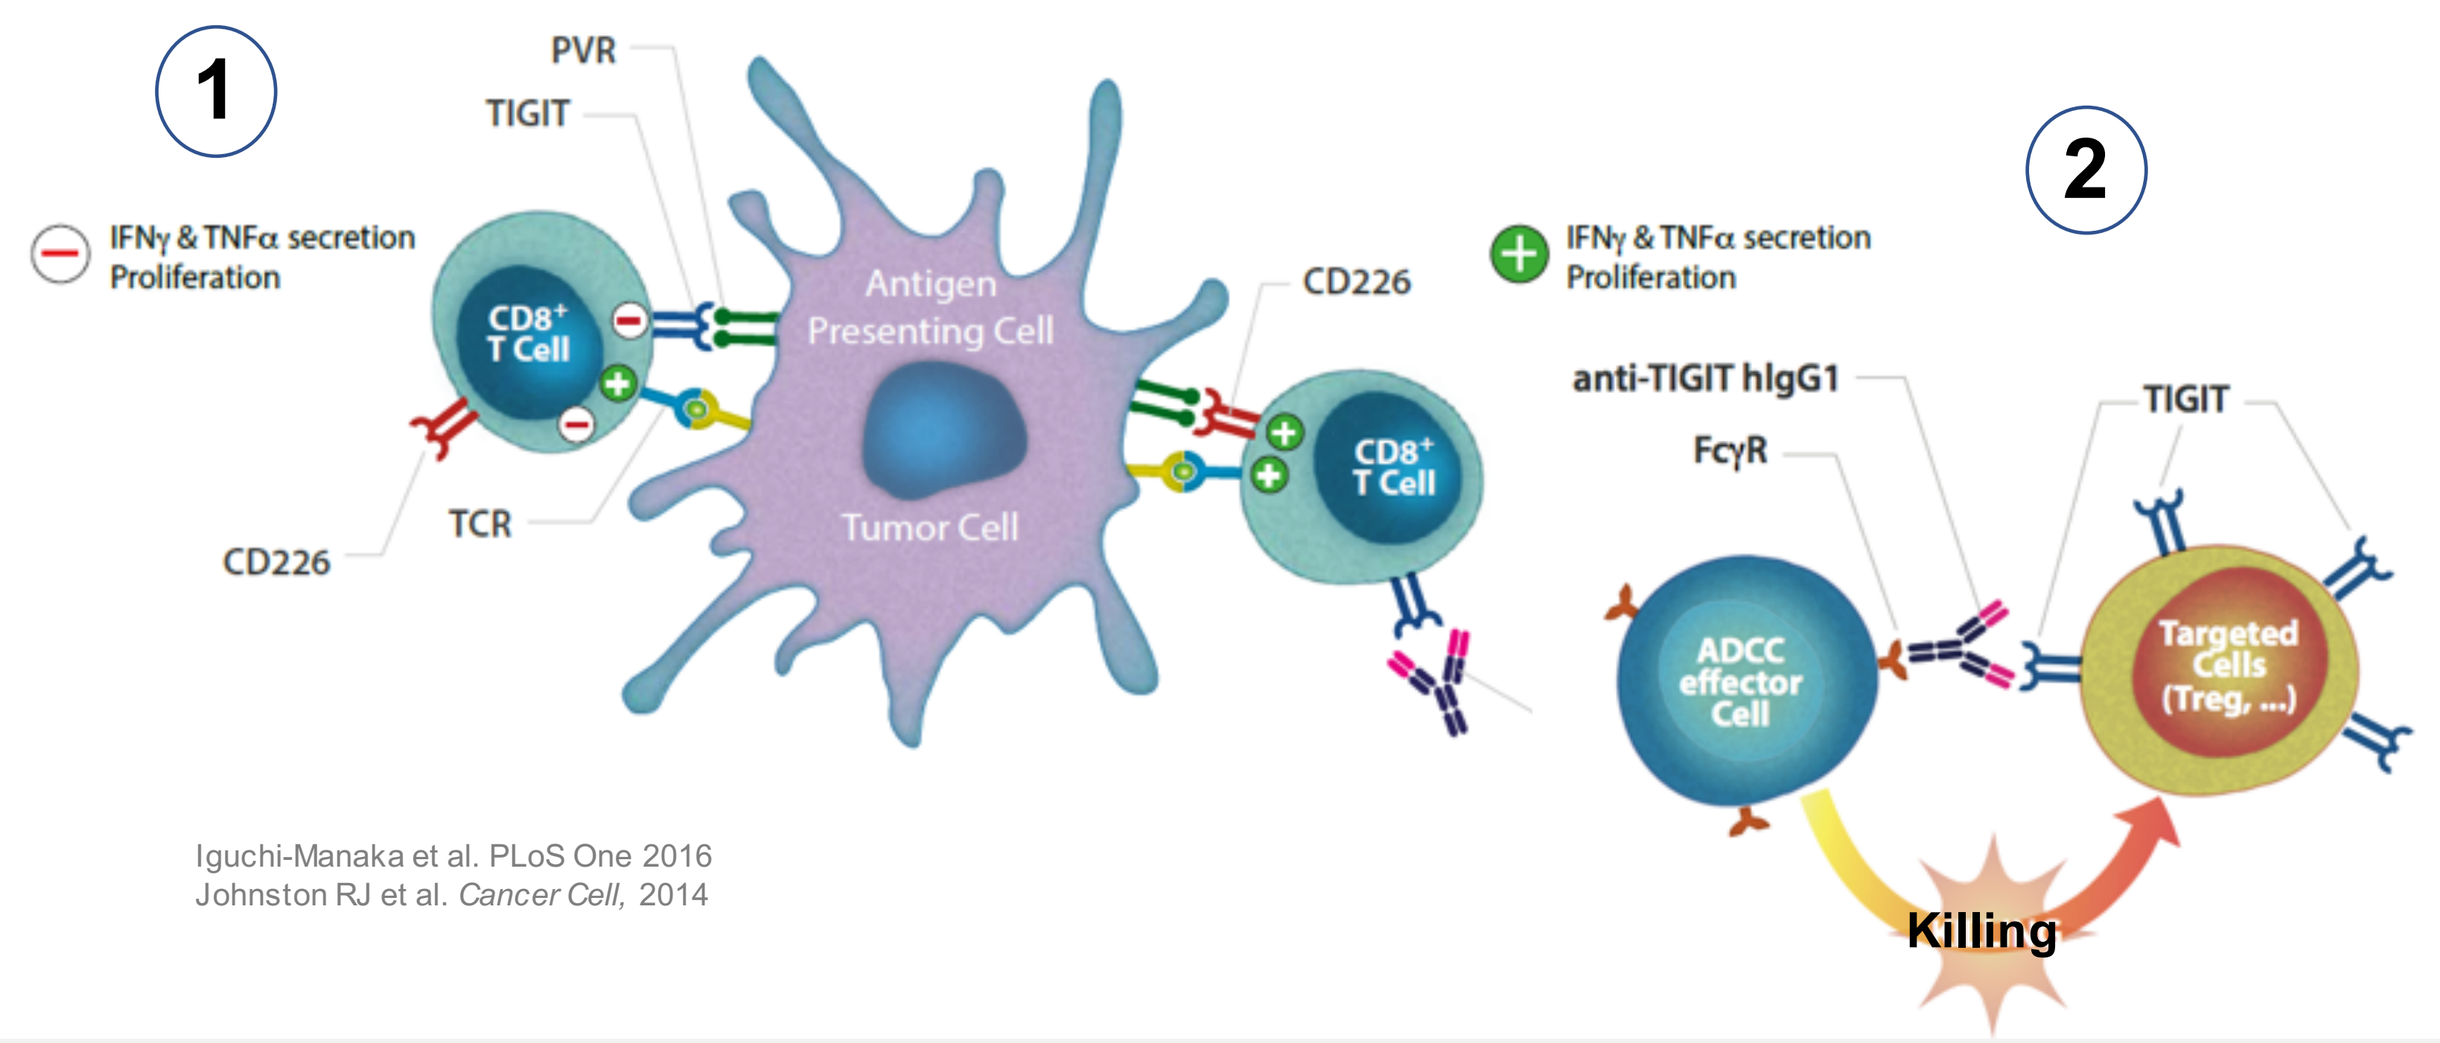
1
PVR
TIGIT
IFNγ & TNFα secretion
Proliferation
CD8⁺ T Cell
Antigen Presenting Cell
Tumor Cell
CD226
TCR
CD226
CD8⁺ T Cell
2
IFNγ & TNFα secretion
Proliferation
anti-TIGIT hIgG1
FcγR
TIGIT
ADCC effector Cell
Targeted Cells (Treg, ...)
Killing
Iguchi-Manaka et al. PLoS One 2016
Johnston RJ et al. Cancer Cell, 2014

1 drug = multiple anti-tumor mechanisms of action

1. Reactivation of immune response by
 - Suppressing TIGIT-mediated inhibitory signaling
 - Increasing ligand availibility for CD226 co-stimulatory receptor
2. Depletion of TIGIT⁺ highly suppressive Treg subpopulation and TIGIT⁺ tumor cells with ADCC-triggering isotype (FcγR dependent)

EOS884448 BINDS WITH HIGH AFFINITY TO HUMAN AND CYNO TIGIT AND PREVENTS LIGAND BINDING TO TIGIT

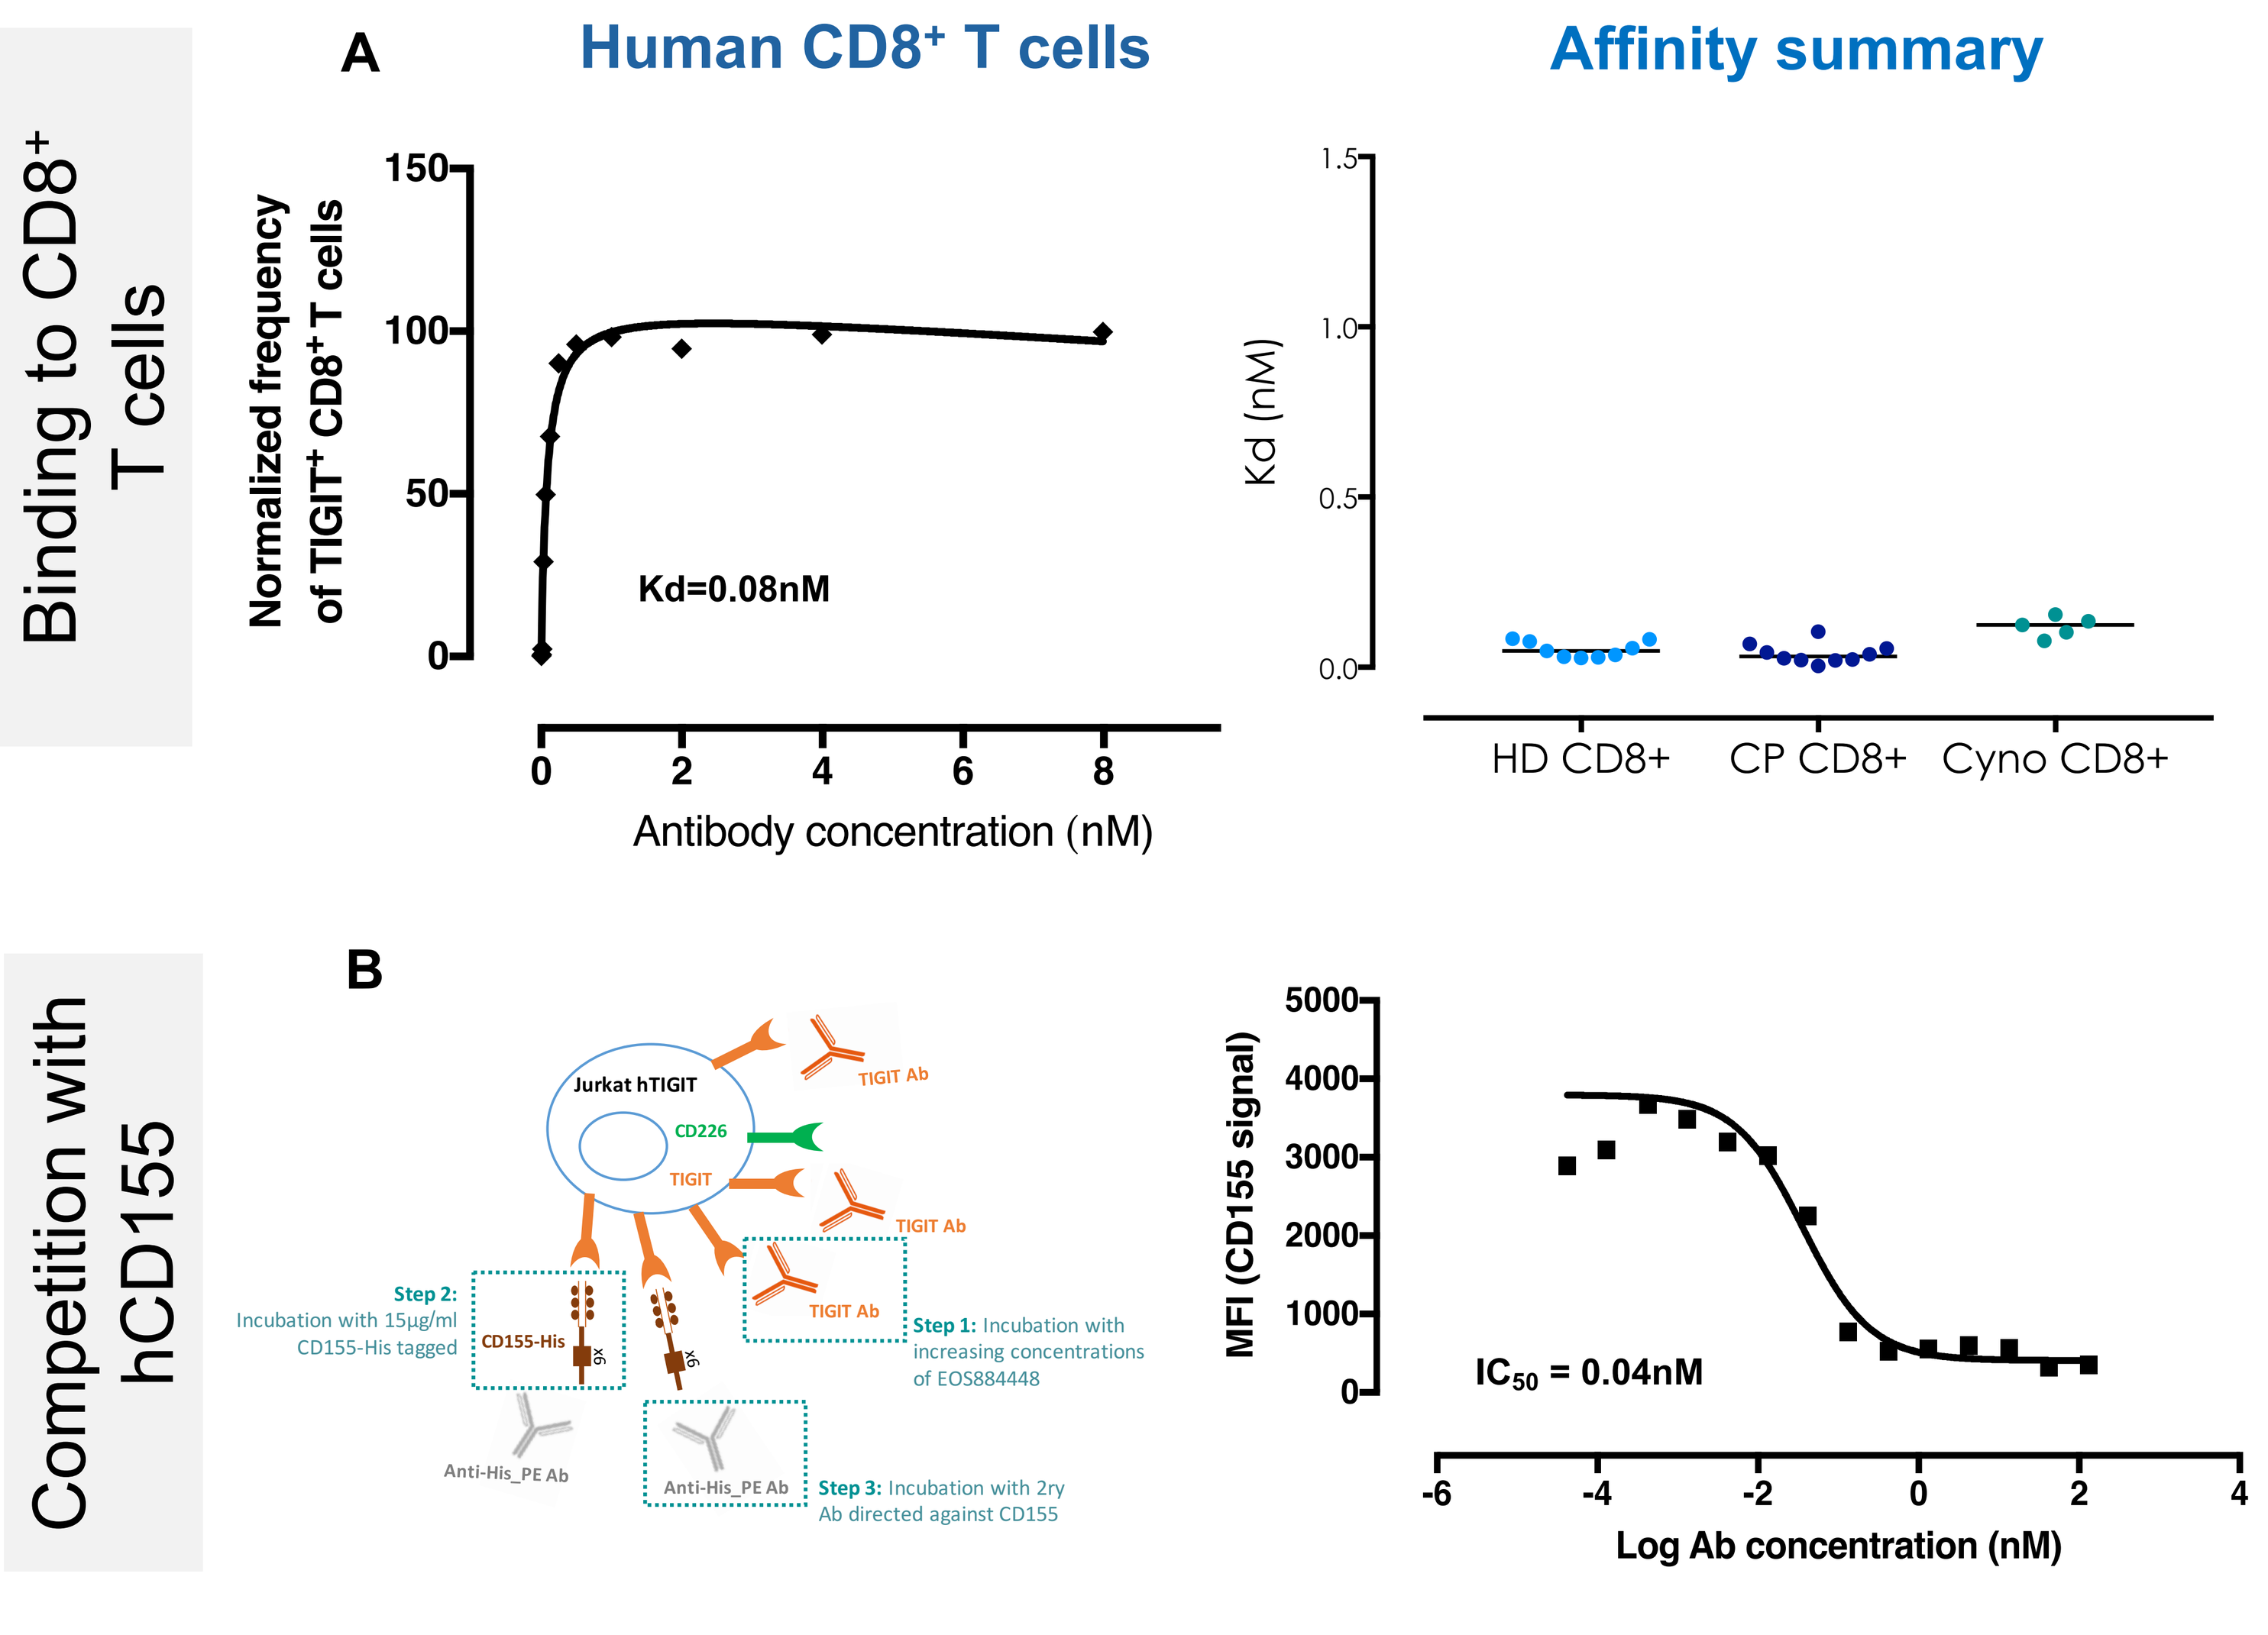
Binding to CD8⁺ T cells
A
Human CD8⁺ T cells
Normalized frequency of TIGIT⁺ CD8⁺ T cells
Kd=0.08nM
Antibody concentration (nM)
Affinity summary
Kd (nM)
HD CD8+
CP CD8+
Cyno CD8+
Competition with hCD155
B
Jurkat hTIGIT
CD226
TIGIT
TIGIT Ab
Step 1: Incubation with increasing concentrations of EOS884448
Step 2: Incubation with 15µg/ml CD155-His tagged
CD155-His
Anti-His_PE Ab
Step 3: Incubation with 2ry Ab directed against CD155
MFI (CD155 signal)
IC_{50} = 0.04nM
Log Ab concentration (nM)

Fig. 1. EOS884448 displays a sub-nM affinity to primary T cells from healthy donors and cancer patients and potently prevents binding of TIGIT ligands. (A) Human PBMC from healthy donors (HD) and cancer patients (CP), as well as cynomolgous PBMC were incubated with dose escalating concentrations of EOS884448. Kd of binding was calculated based on normalized frequency of TIGIT⁺CD8⁺ at the different concentrations tested. (B) EOS884448 prevents CD155 ligand binding to TIGIT overexpressing Jurkat cells, showing IC50 values at sub-nM range.

EOS884448 POTENTLY PREVENTS TIGIT INHIBITORY SIGNALING IN PRESENCE OF CD155 LIGAND

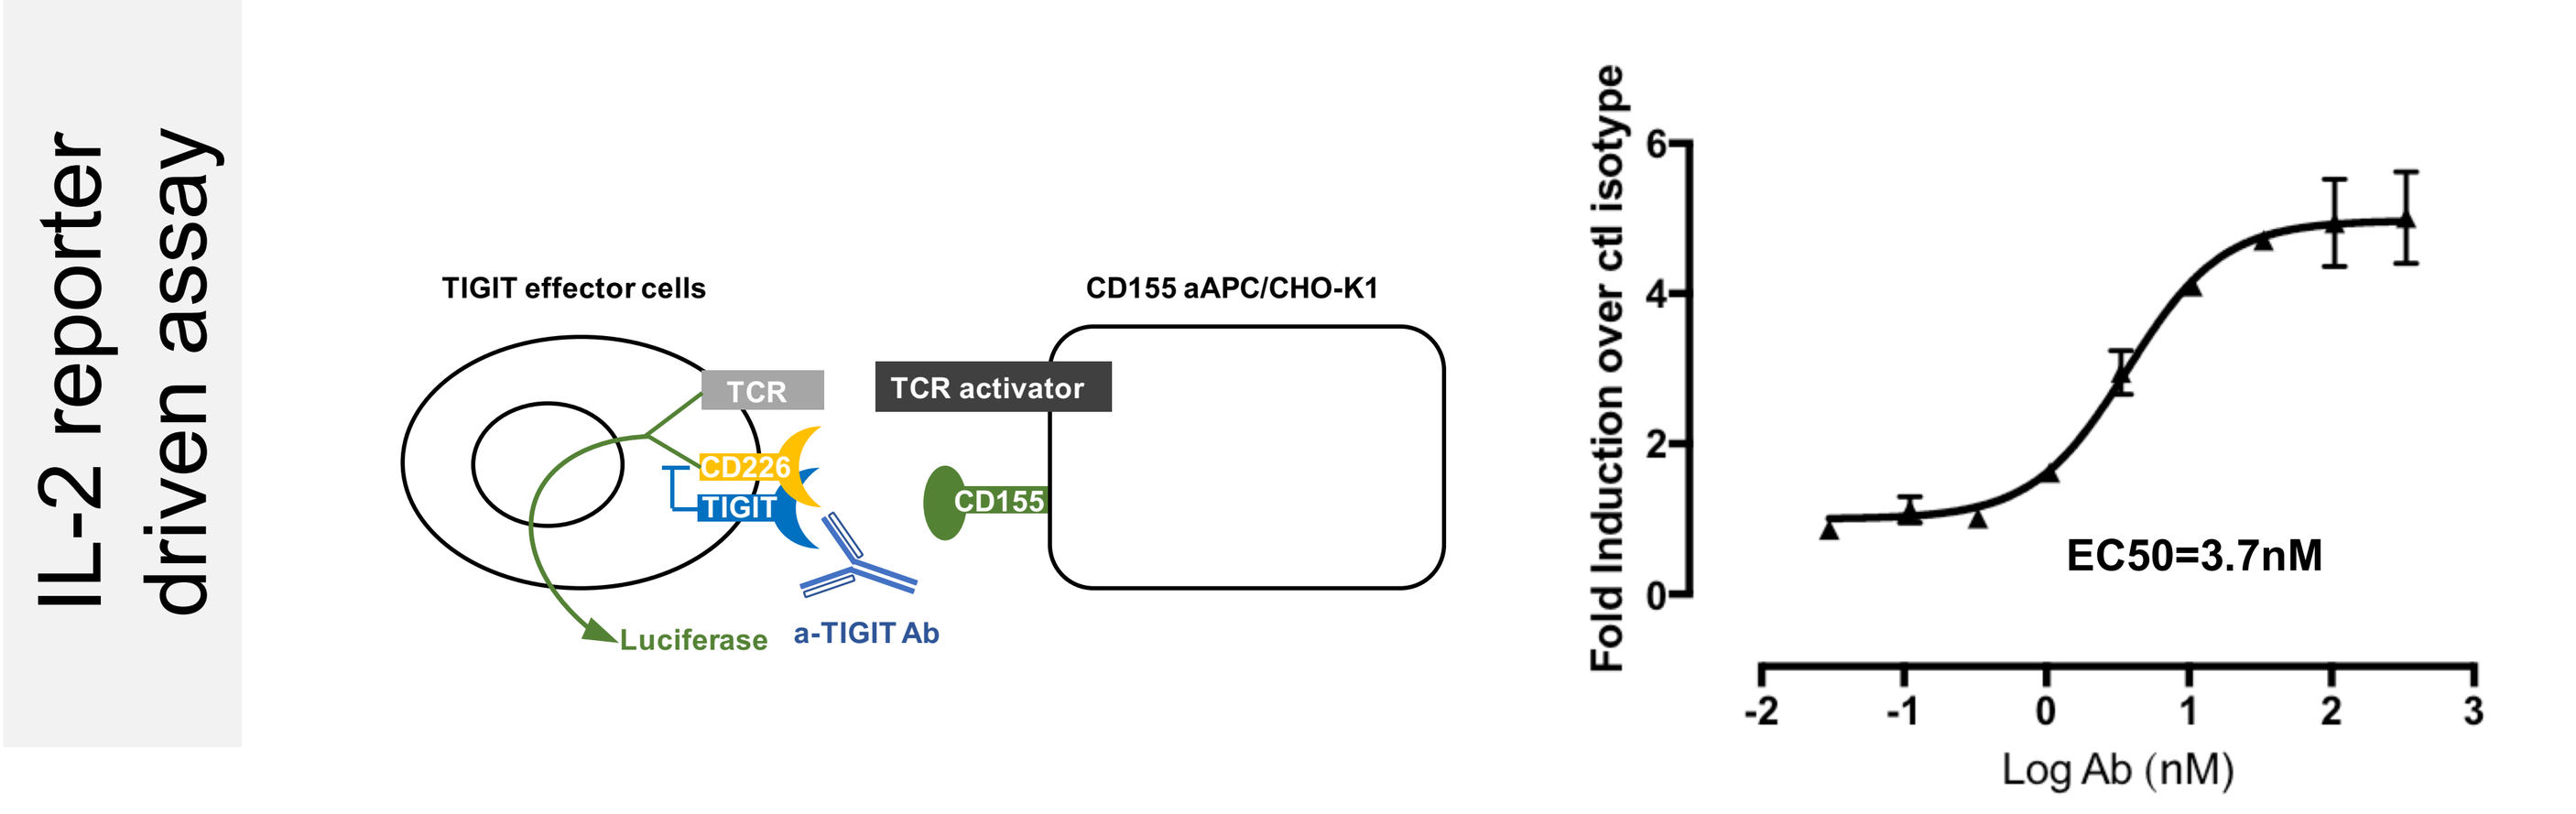
IL-2 reporter driven assay
TIGIT effector cells
CD155 aAPC/CHO-K1
TCR
TCR activator
CD226
TIGIT
CD155
Luciferase
a-TIGIT Ab
Fold Induction over ctl isotype
EC50=3.7nM
Log Ab (nM)

Fig. 2. EOS884448 potently blocks TIGIT-CD155 mediated immunosuppression. Jurkat T cells expressing human TIGIT with a luciferase reporter driven by a native promoter that can respond to both TCR activation and CD226 co-stimulation were stimulated with APC-like cells expressing CD155 (CHO-TCR-CD155). Addition of EOS884448 during stimulation prevents TIGIT:CD155 induced inhibition and increases T cell activation-mediated luminescence.

EOS884448 INCREASES IFNγ SECRETION IN T CELLS FROM HEALTHY AND CANCER PATIENTS

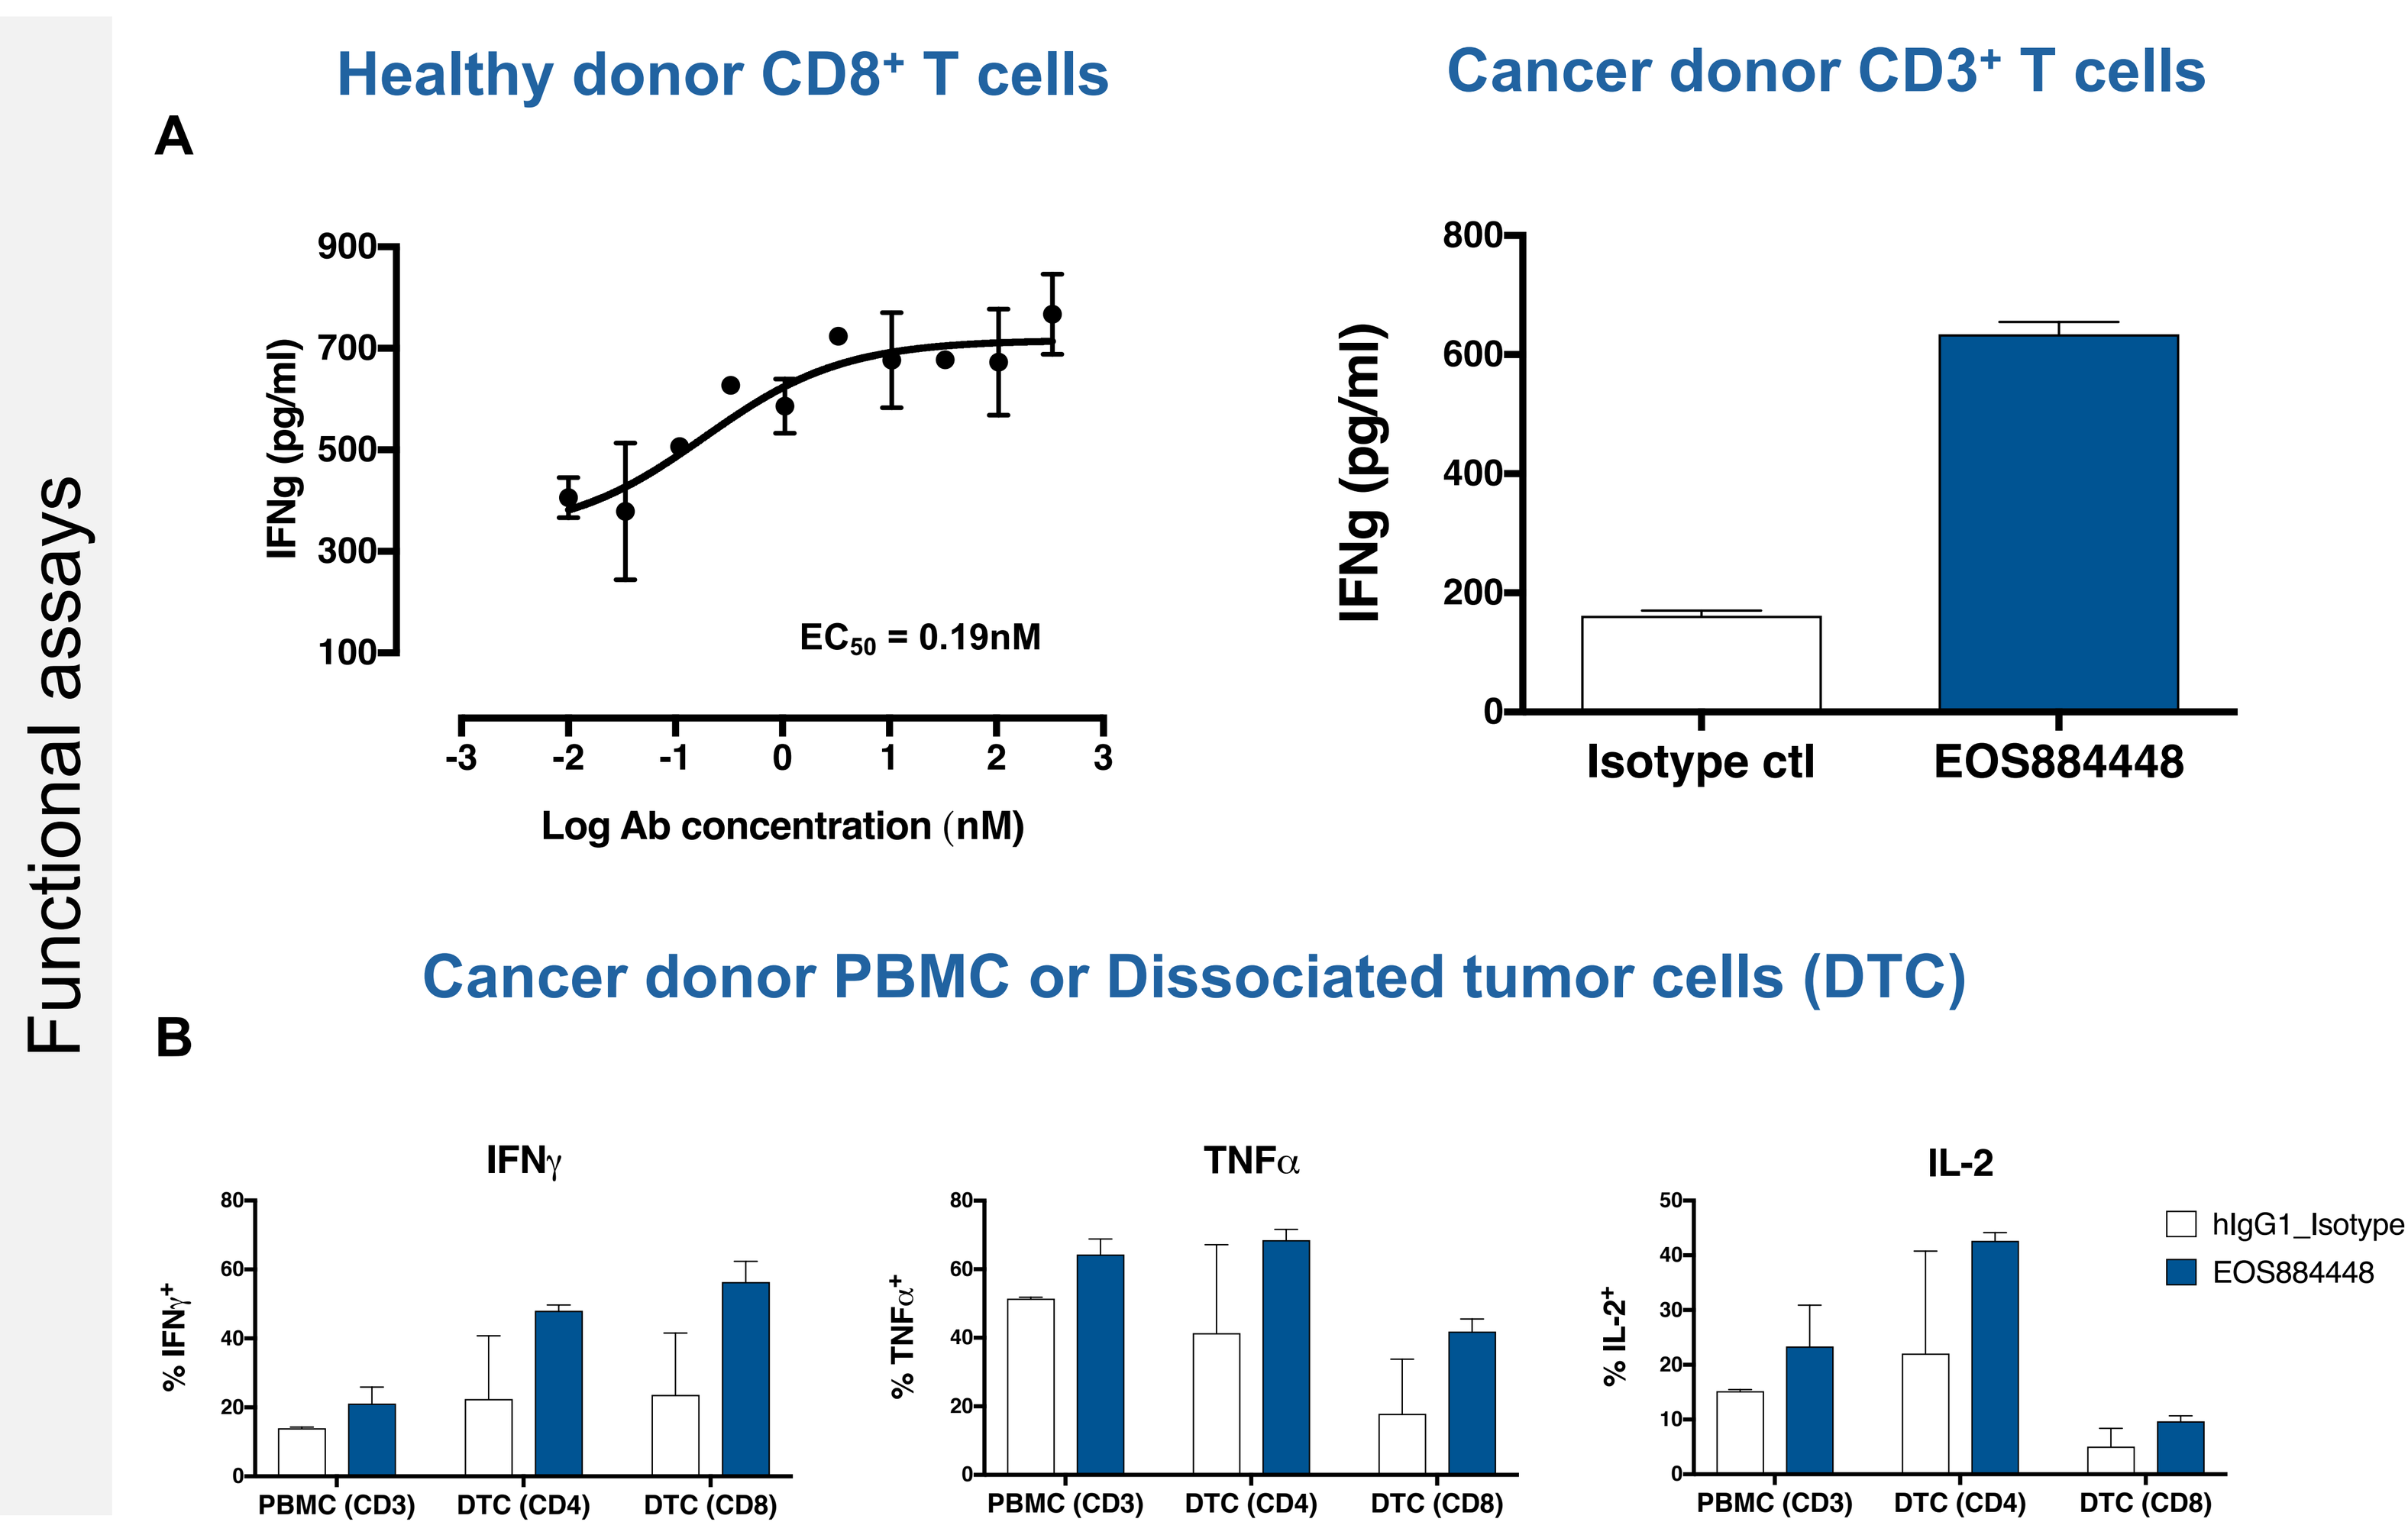
Functional assays
A
Healthy donor CD8⁺ T cells
IFNg (pg/ml)
EC_{50} = 0.19nM
Log Ab concentration (nM)
Cancer donor CD3⁺ T cells
IFNg (pg/ml)
Isotype ctl
EOS884448
B
Cancer donor PBMC or Dissociated tumor cells (DTC)
IFNγ
TNFα
IL-2
% IFNγ⁺
% TNFα⁺
% IL-2⁺
PBMC (CD3)
DTC (CD4)
DTC (CD8)
hIgG1_Isotype
EOS884448

Fig. 3. EOS884448 potently restores pro-inflammatory cytokines in presence of CD155 ligand. (A) Human primary CD3⁺ or CD8⁺ T cells were stimulated with APC-like cells (CHO-TCR-CD155). Addition of aTIGIT during stimulation prevents CD155-induced inhibition and increases IFNγ production in T cells from healthy donors and cancer patients. (B) EOS884448 restores cytokine release from freshly isolated cancer patient PBMC or tumor infiltrated lymphocytes present among dissociated tumor when incubated overnight with aCD3/CD28 stimulation, as measured by intracellular staining.

EOS884448 POSITIVELY MODULATES ACTIVITY OF NON-CONVENTIONAL $\gamma\delta$ T CELLS

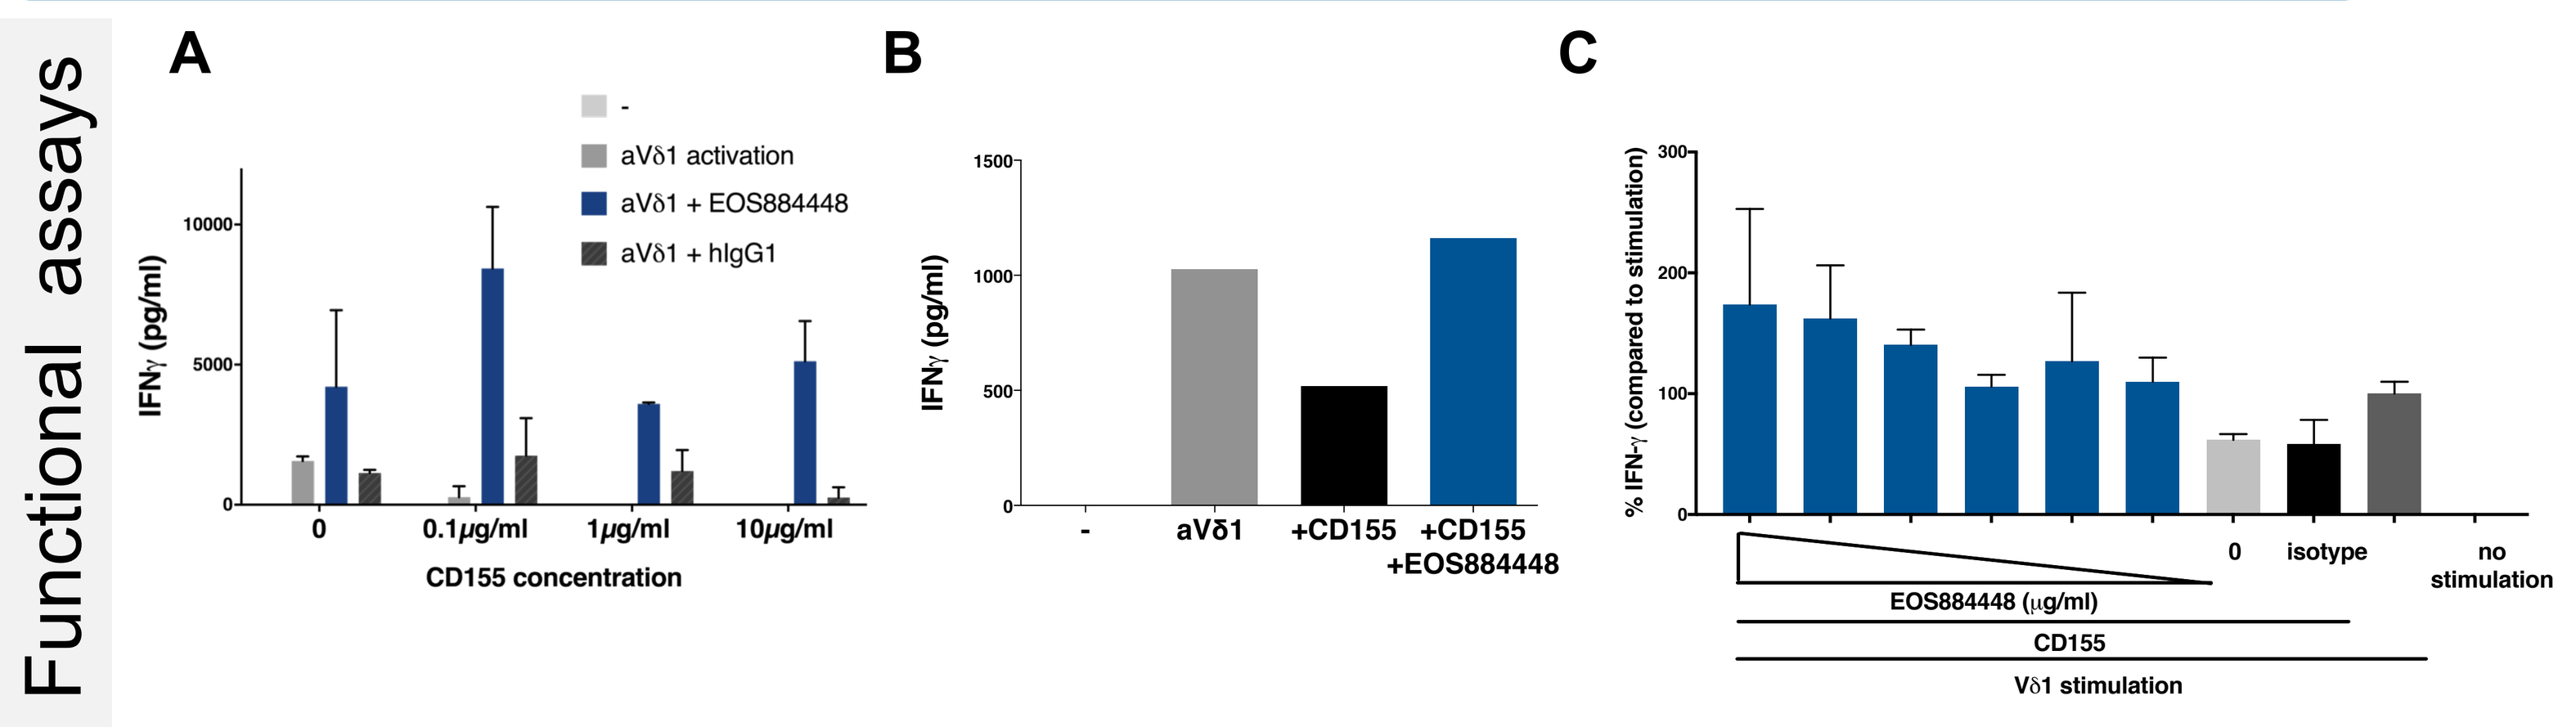
Functional assays
A
-
aVδ1 activation
aVδ1 + EOS884448
aVδ1 + hIgG1
IFNγ (pg/ml)
0
0.1µg/ml
1µg/ml
10µg/ml
CD155 concentration
B
IFNγ (pg/ml)
-
aVδ1
+CD155
+CD155 +EOS884448
C
% IFN-γ (compared to stimulation)
0
isotype
no stimulation
EOS884448 (µg/ml)
CD155
Vδ1 stimulation

Fig. 4. EOS884448 restores IFN-γ production by $\gamma\delta$ T cells. Magnetically isolated Vδ1⁺ $\gamma\delta$ T cells (A) or PBMC (B) from CMV⁺ donors were activated with anti-Vδ1 and IL-15. When recombinant CD155 was added to the culture, IFN-γ production was reduced in a dose-dependant manner and EOS884448 was able to prevent this inhibition. Treatment with EOS884448 increases the secretion of IFN-γ at a level superior than the control without CD155. (C) Dose-dependant increase in IFN-γ production by EOS884448 on total PBMC activated with anti-Vδ1 and IL-15.

EOS884448 PREFERENTIALLY DEPLETES TREG IN HUMAN PBMC FROM HEALTHY AND CANCER DONORS

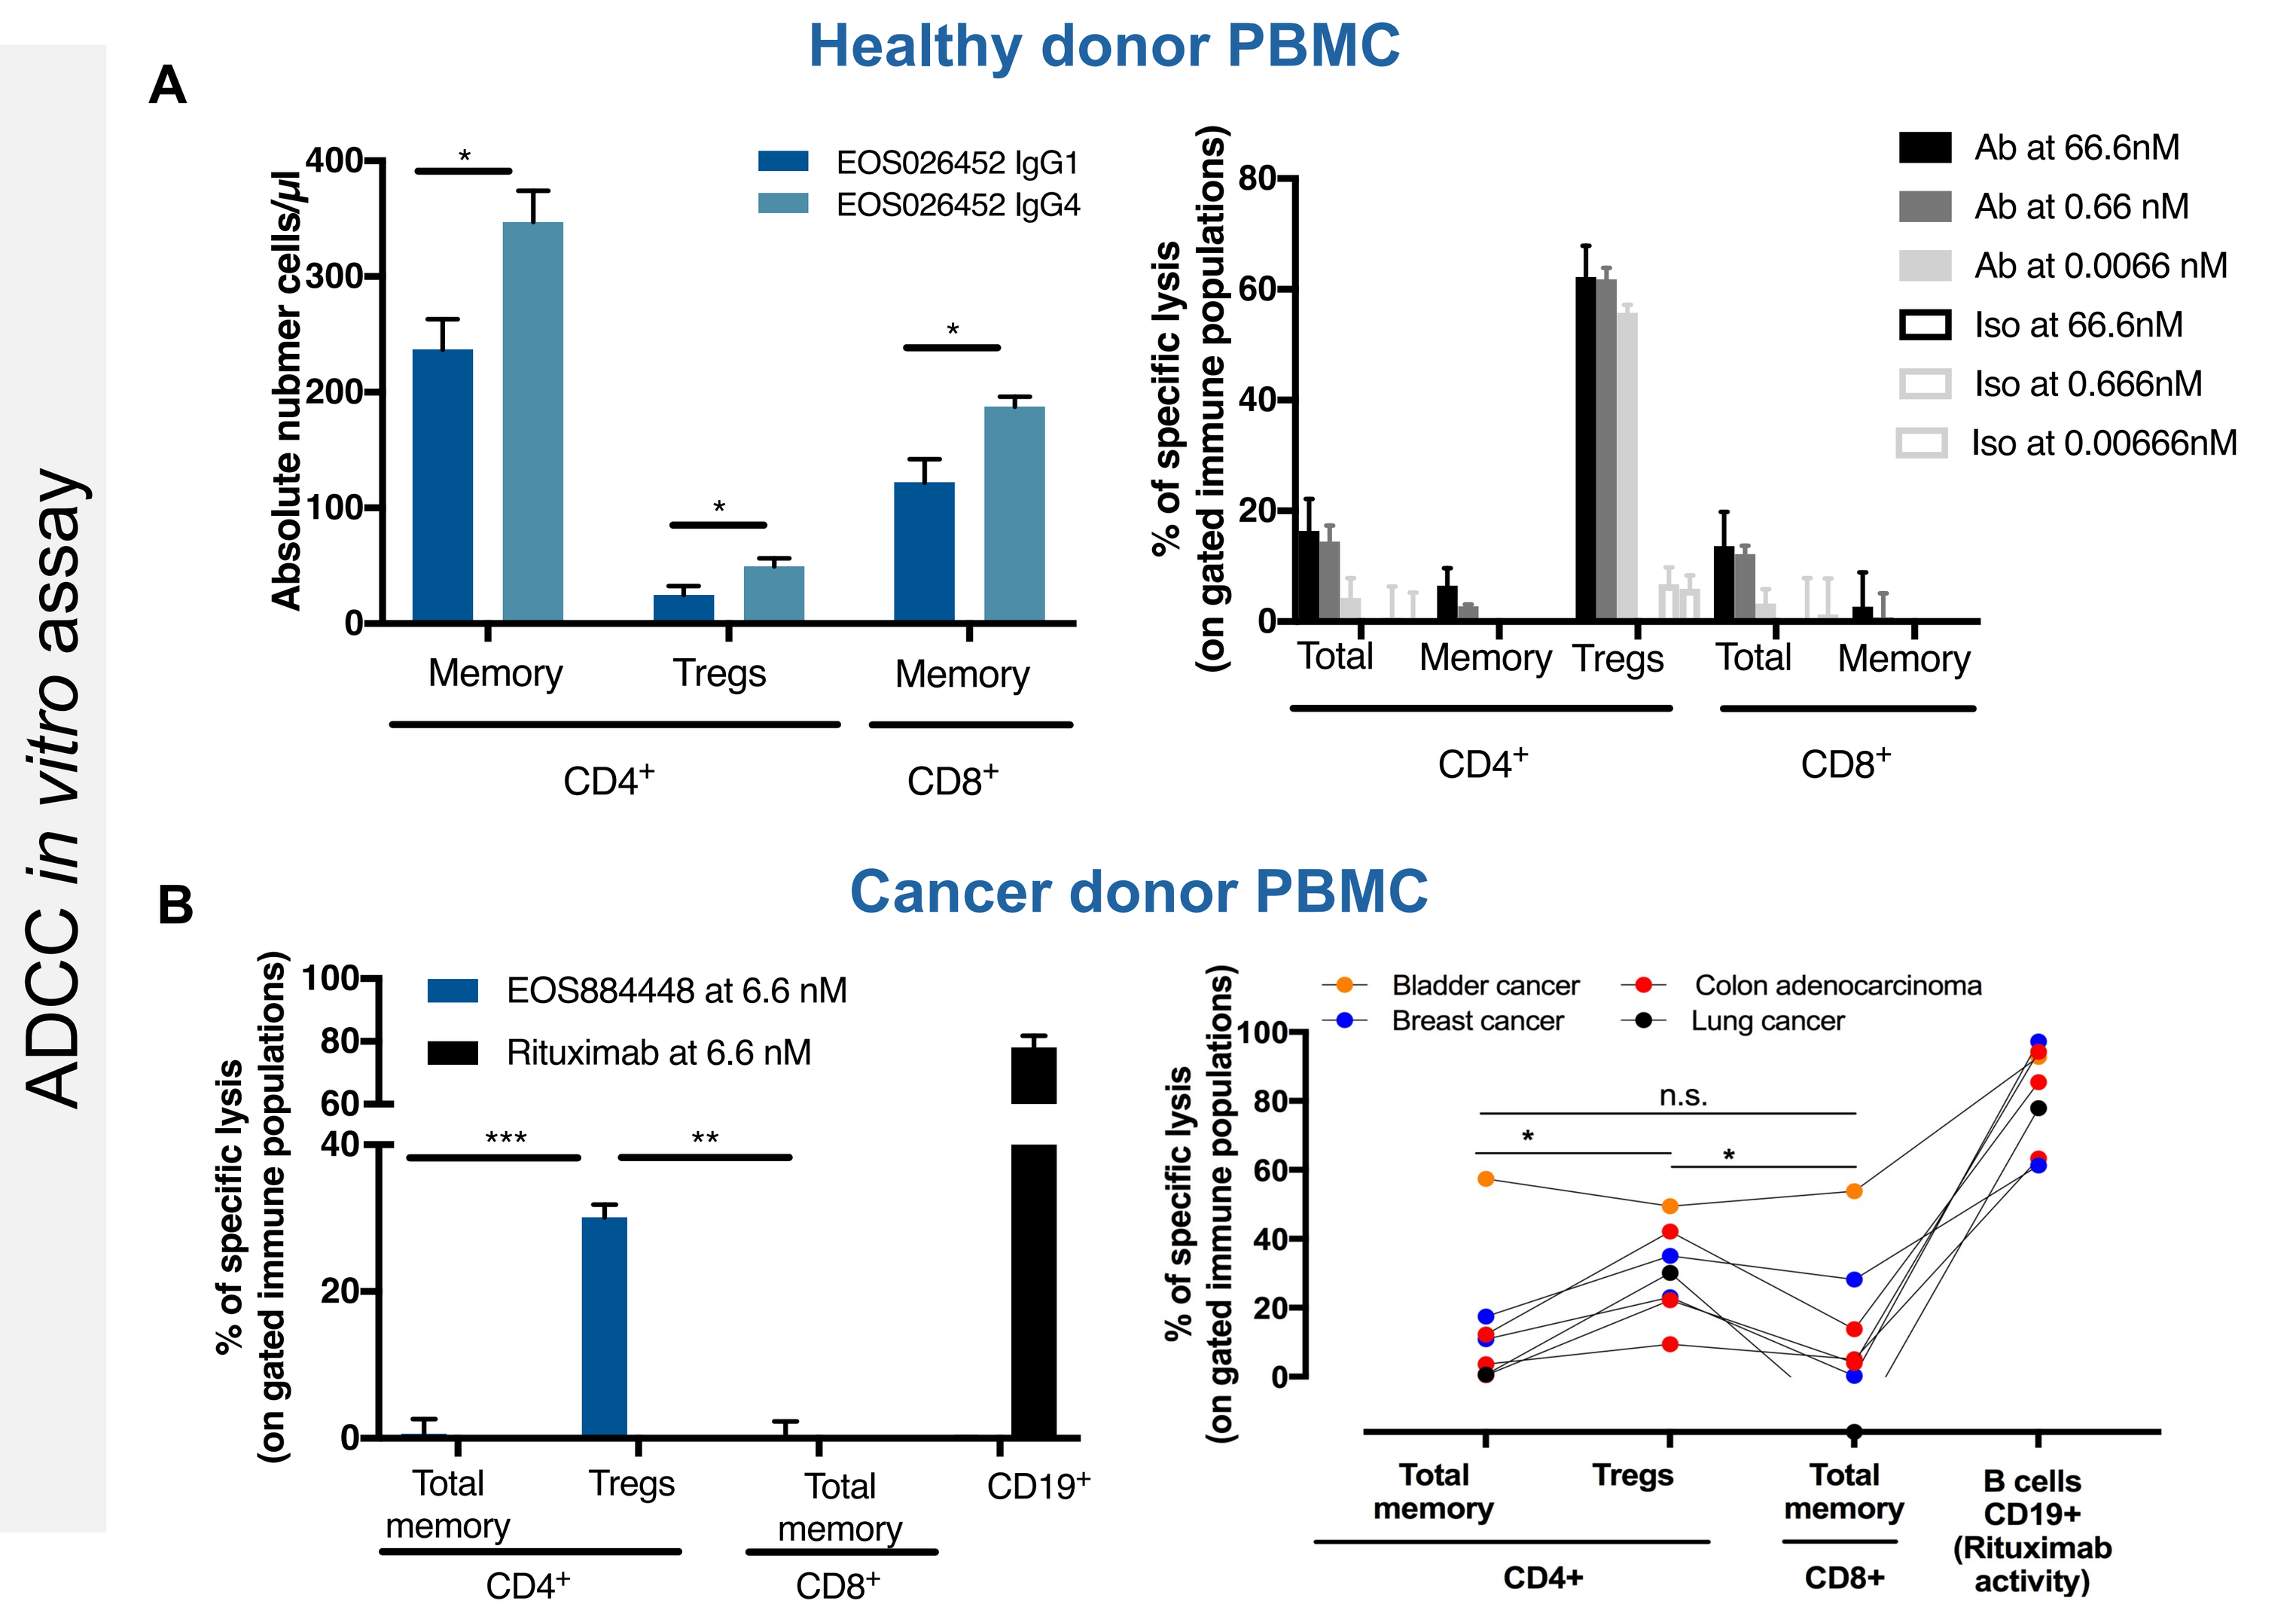
ADCC in vitro assay
A
Healthy donor PBMC
Absolute nubmer cells/µl
EOS026452 IgG1
EOS026452 IgG4
Memory
Tregs
Memory
CD4⁺
CD8⁺
% of specific lysis (on gated immune populations)
Ab at 66.6nM
Ab at 0.66 nM
Ab at 0.0066 nM
Iso at 66.6nM
Iso at 0.666nM
Iso at 0.00666nM
Total
Memory
Tregs
Total
Memory
B
Cancer donor PBMC
EOS884448 at 6.6 nM
Rituximab at 6.6 nM
Total memory
Tregs
Total memory
CD19⁺
Bladder cancer
Breast cancer
Colon adenocarcinoma
Lung cancer
n.s.
B cells CD19+ (Rituximab activity)
CD4+
CD8+

Fig. 5. EOS884448, as an hIgG1, shows preferential depletion of Tregs over memory CD4⁺ and CD8⁺ T cells. (A) Absolute cell counts and depletion of different T cell populations expressing TIGIT is shown when PBMC are incubated *in vitro* in presence of EOS026452 (EOS884448 parent) for 20h and quantified by FACS. EOS026452 shows a preferential depletion of Treg cells at 66.6 nM or lower concentrations in healthy donors. (B) Similar preferential depletion with EOS884448 is observed in PBMC from cancer patients suffering from different solid tumor types (*p<0.05).

CONCLUSIONS

EOS884448 is a highly potent antagonist aTIGIT mAb that:

- ❖ Binds with high affinity to human and cynomolgous TIGIT⁺ T cells.
- ❖ Competes for binding with TIGIT ligands.
- ❖ Prevents CD155-TIGIT mediated immunosuppression and restores effector T cell function.

⇒ All at sub-nM concentrations

- ❖ Preferentially depletes human Tregs *in vitro.*
- ❖ **Preclinical data support the transition of EOS884448 to the clinic as a potent a-TIGIT antagonist Ab.**

iTeos Therapeutics, Gosselies, Belgium and Cambridge, MA, USA

Corresponding author: gregory.driessens@iteostherapeutics.com

Wallonie | SPW Service public de Wallonie | Avec le soutien de la DGO6 Département du Développement Technologique