Development and validation of a medium-throughput phenotypic screening platform for the identification of novel immuno-oncology targets

Léa Legrand, Marie-Claire Letellier, Ariane Scoumanne, João Marchante, Virginie Rabolli, Murielle Martini, Stefano Crosignani, Christophe Quéva, Michel Detheux, Sandra Cauwenberghs, Jakub M. Swiercz

With support from DG06
Département du Développement Technologique

ABSTRACT

Immunotherapy is a major breakthrough in cancer treatment. Although recent strategies, including immune-checkpoint blockade have demonstrated therapeutic benefit in patients with various advanced cancers, further understanding of human immunopathology, particularly in the tumor microenvironment, is essential to improve these therapeutic approaches.

In order to shed light on novel immune suppressive mechanisms in tumor, iTeos Therapeutics has developed a target discovery and drug repurposing platform based on phenotypic screening assays. We have established a co-culture assay combining human tumor immune suppressive cells and T-cells. This assay is flexible to allow the screening of chemogenomics, shRNA and cDNA libraries. Multi-parameter readouts are combined to assess both T cell activation and proliferation, through high-content imaging of T cell cluster formation complemented with detection of IFNγ secretion, and tumor cell death, as assessed using a cytotoxicity assay. The 96-well format of the assay allows medium-throughput testing of up to 3000 samples/screen and his robust to determine EC_{50}, potency ranking and target deconvolution.

We report here a proof-of-concept study in which we evaluated the ability to detect metabolic immune-oncology targets in A549 cells, a lung cancer immune suppressive cell line. A549 express indoleamine-2,3-dioxygenase 1 (IDO1), an enzyme overexpressed in cancer that mediates local T-cell suppression through depleting the essential amino acid tryptophan. The assay conditions were validated with an IDO1 inhibitor as positive control and subsequently scaled up for automation. A commercially available small molecule library of 1900 compounds, with a high percentage of clinically tested drugs was screened. The library was tested at two different concentrations (0.3μM and 3μM), with two independent T-cell donors and spiked with IDO1 inhibitor as control. Combined analysis of T-cell activity and tumor killing led to the identification of 42 compounds with activity on multiple potential immune suppressive pathways, including metabolism, epigenetics, autophagy, TGFβ, Wnt/β-catenin and TNFα/NF-κB signaling.

EXPERIMENTAL SETUP

AIM OF THE STUDY:
Design and validate robust system allowing identification of novel modulators of tumor-mediated immunosuppression

Choosing compound library:
(Selleck, 1902 compounds) and screening concentration (300nM and 3μM).
Re-formatting of plates using EpMotion

Workflow optimisation and validation

Readouts:
- Clusters acquisition (Incucyte)
- INFγ measurement (AlphaLISA)
- Cell death measurement (CytoToxGlo)

Results analysis (R script and Prism)

Hit identification

Representation of 5-day workflow

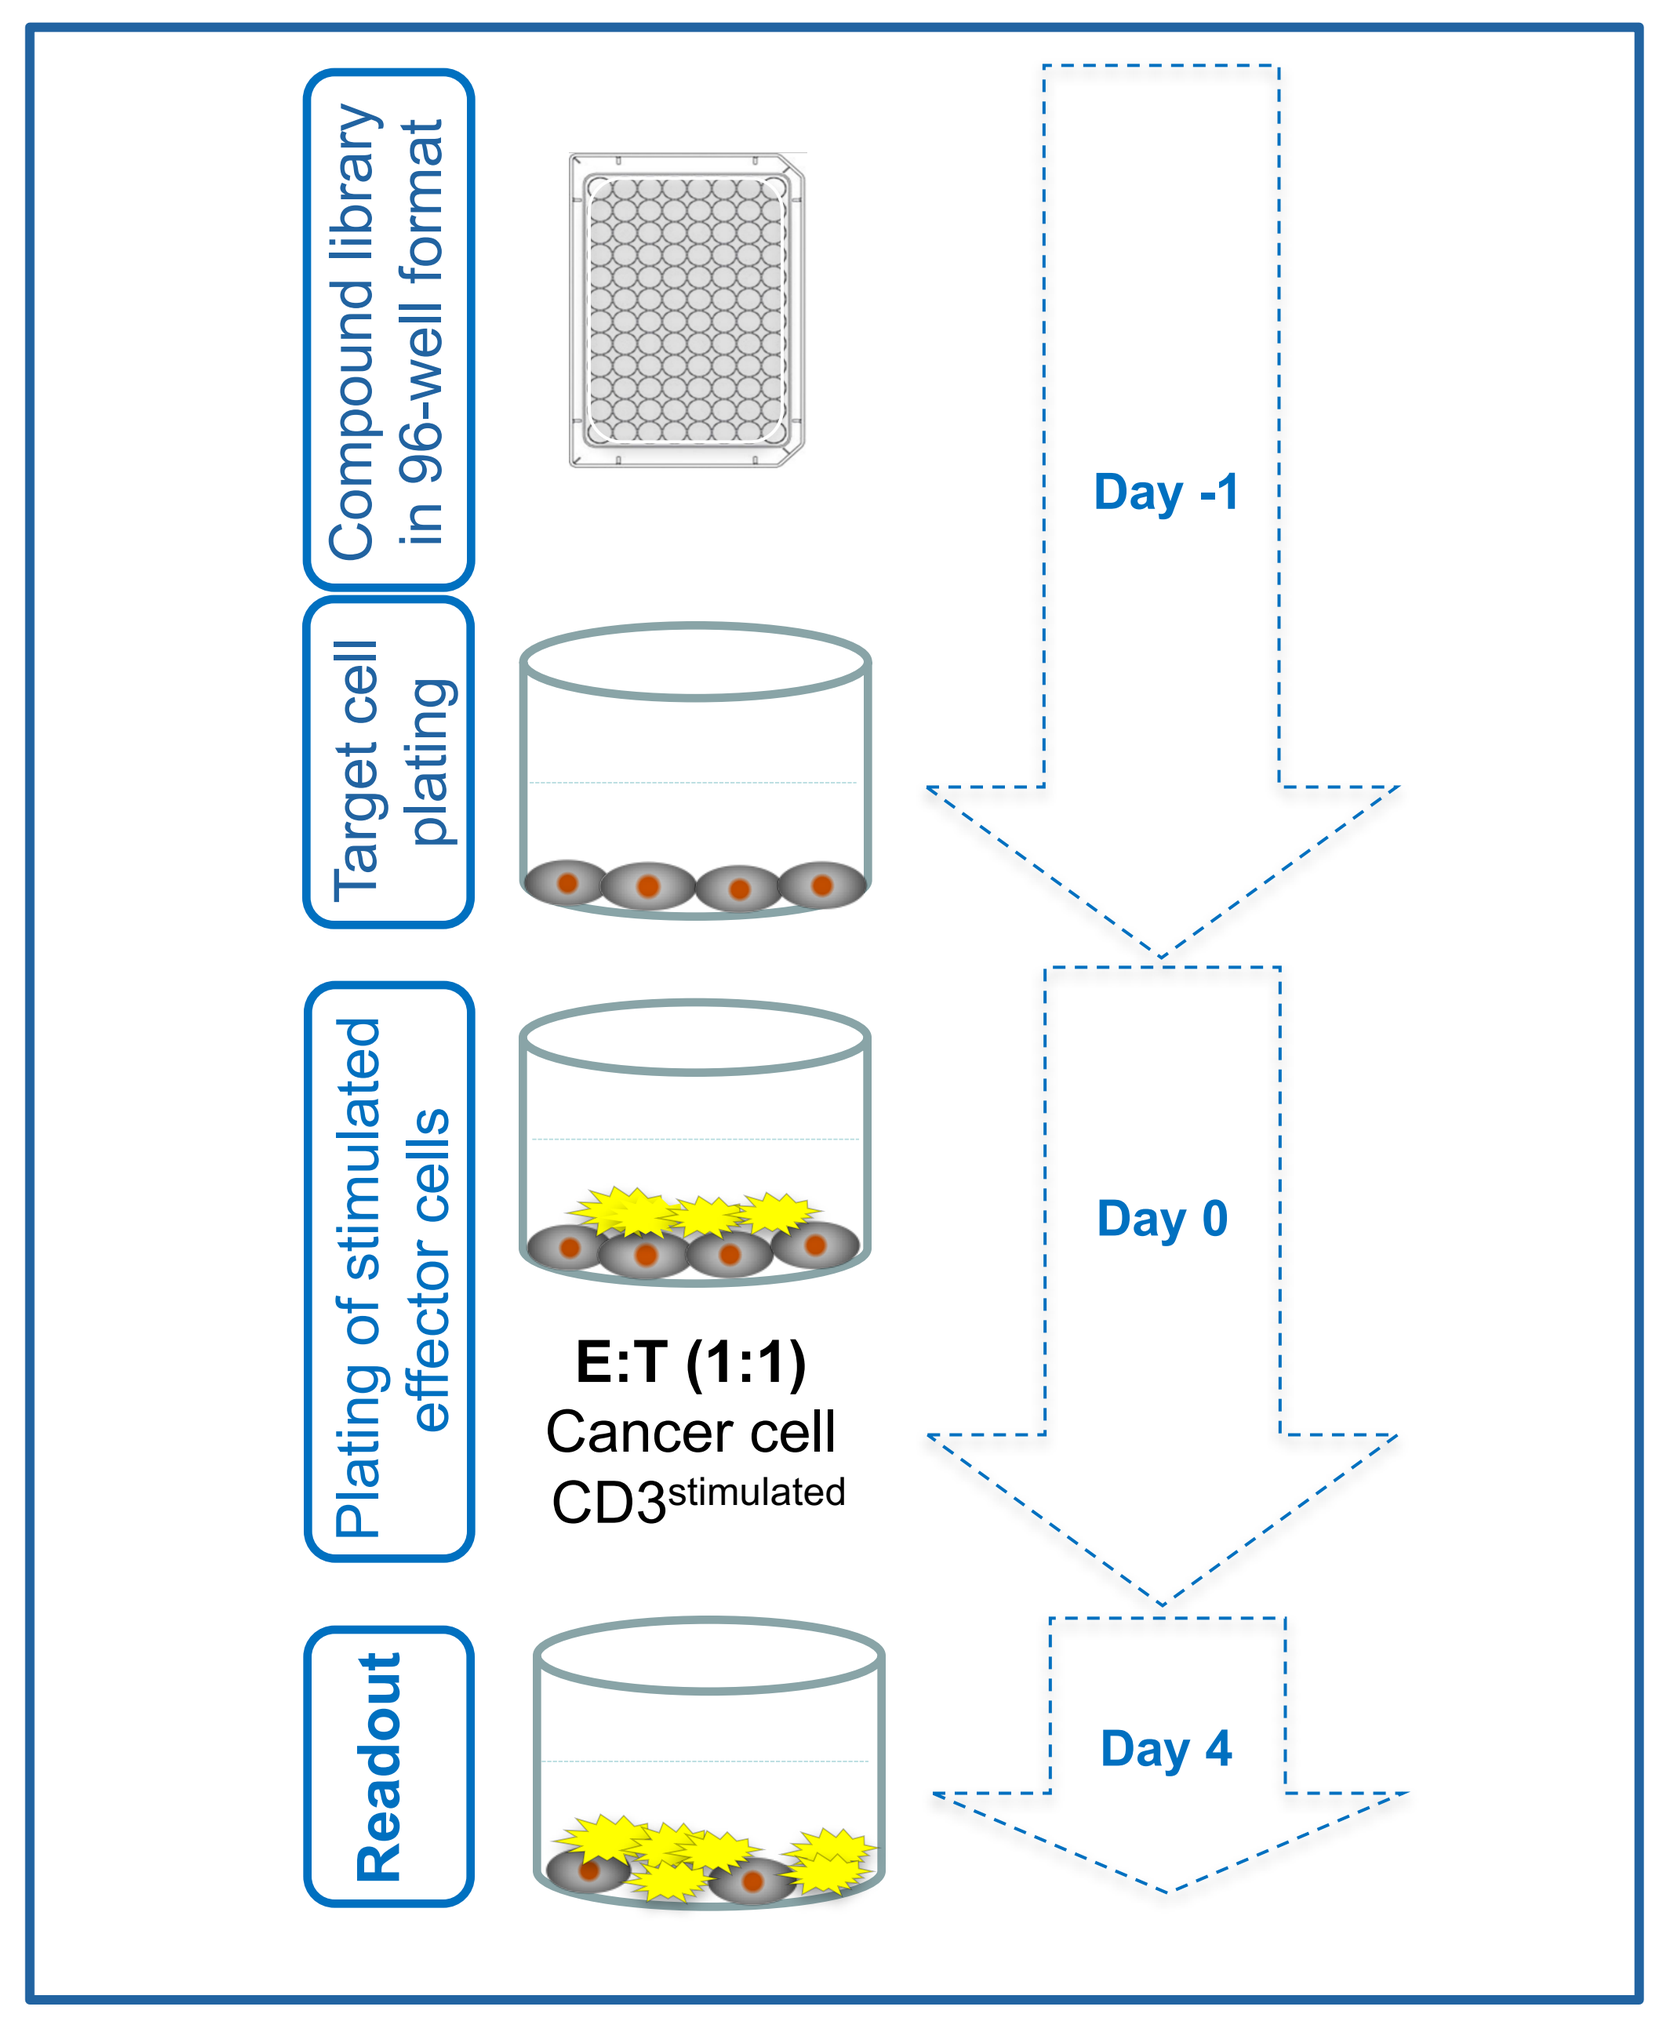
Compound library in 96-well format
Day -1
Target cell plating
Plating of stimulated effector cells
E:T (1:1)
Cancer cell
CD3stimulated
Day 0
Readout
Day 4

READOUT VALIDATION

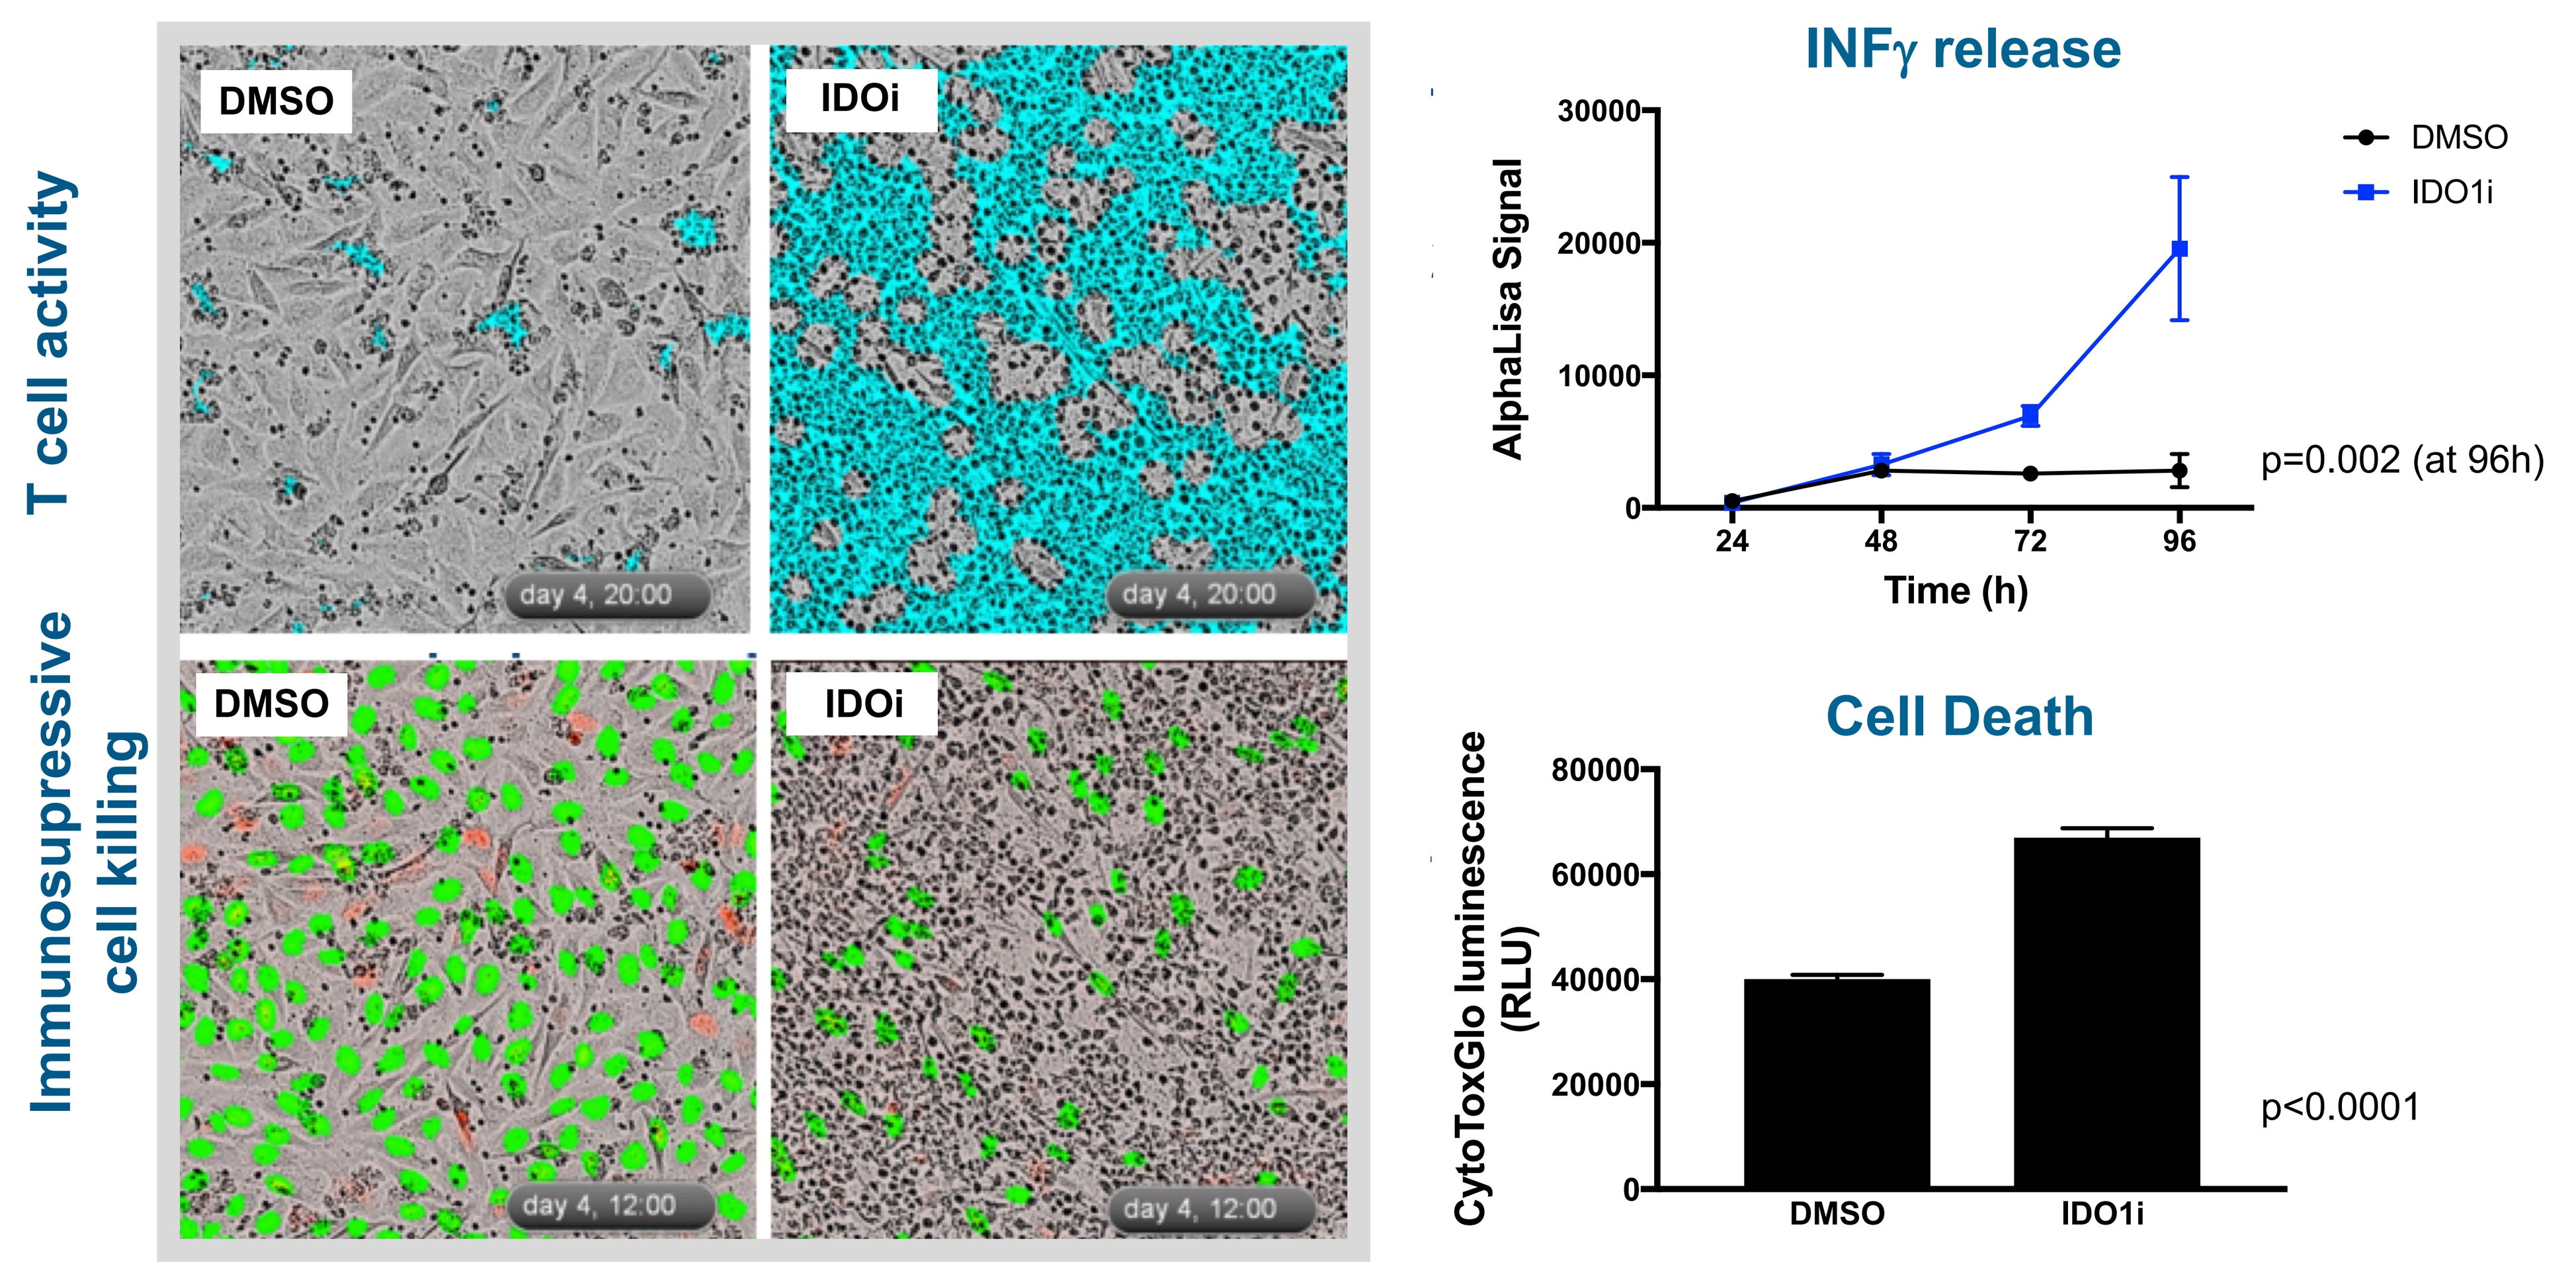
T cell activity
Immunosupressive cell killing
DMSO
IDOi
day 4, 20:00
day 4, 12:00
INFγ release
AlphaLisa Signal
Time (h)
DMSO
IDO1i
p=0.002 (at 96h)
Cell Death
CytoToxGlo luminescence (RLU)
DMSO
IDO1i
p<0.0001

Readout validation at the end of co-culture of A549-RFP cells and CD3+ T cells. (left panel) At day 4 of the co-culture, images were taken using IncuCyte (Essen BioScience) at 10X resolution analyzed using the IncuCyte ZOOM software. T-cell proliferation was assessed using the mask developed at iTeos (in blue) and the nuclei of the A549-RFP cells were visualized in green. Shown are representative images of each condition, showing reversion of T-cell suppression (upper panel) and increased tumor cell killing (lower panel) upon inhibition of the metabolic enzyme IDO1 (IDO1i). (right panel) Supernatants from the A549-RFP cells/CD3+ T cells co-culture were collected tested for INFγ secretion and cell death. Shown are representative plots of both parameters.

HIT IDENTIFICATION STRATEGY

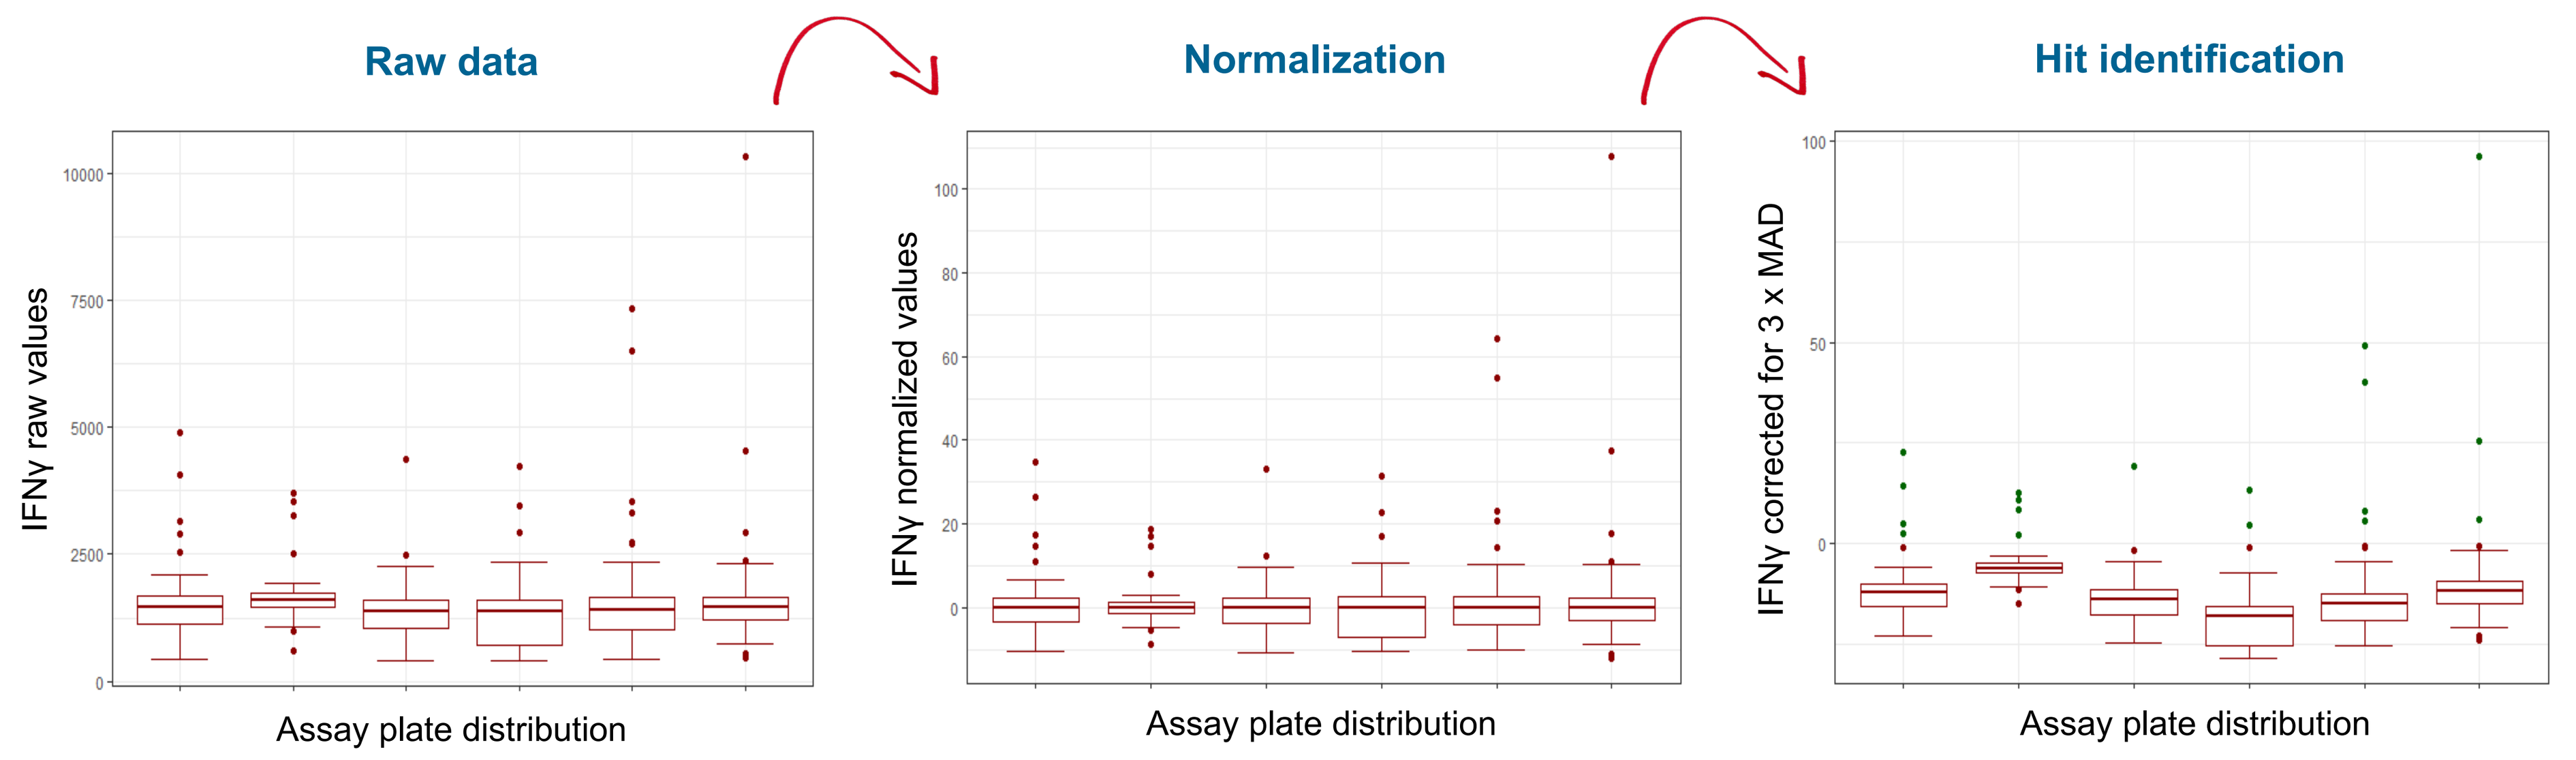
Raw data
Normalization
Hit identification
IFNγ raw values
IFNγ normalized values
IFNγ corrected for 3 x MAD
Assay plate distribution

IFNγ example for the hit identification strategy. Raw values from the different plates were normalized by using the median of DMSO control as 0% and the IC_{90} of the IDO1 compound as 100%. Hit identification was done by subtracting the value of 3x Median Absolute Deviation (MAD) from each plate to each individual value of the same plate. This strategy of normalizing and then subtracting the 3xMAD, although heavily stringent, it allows a larger confidence on the hit identification and, at the same time, the ranking of the compounds.

HIT IDENTIFICATION

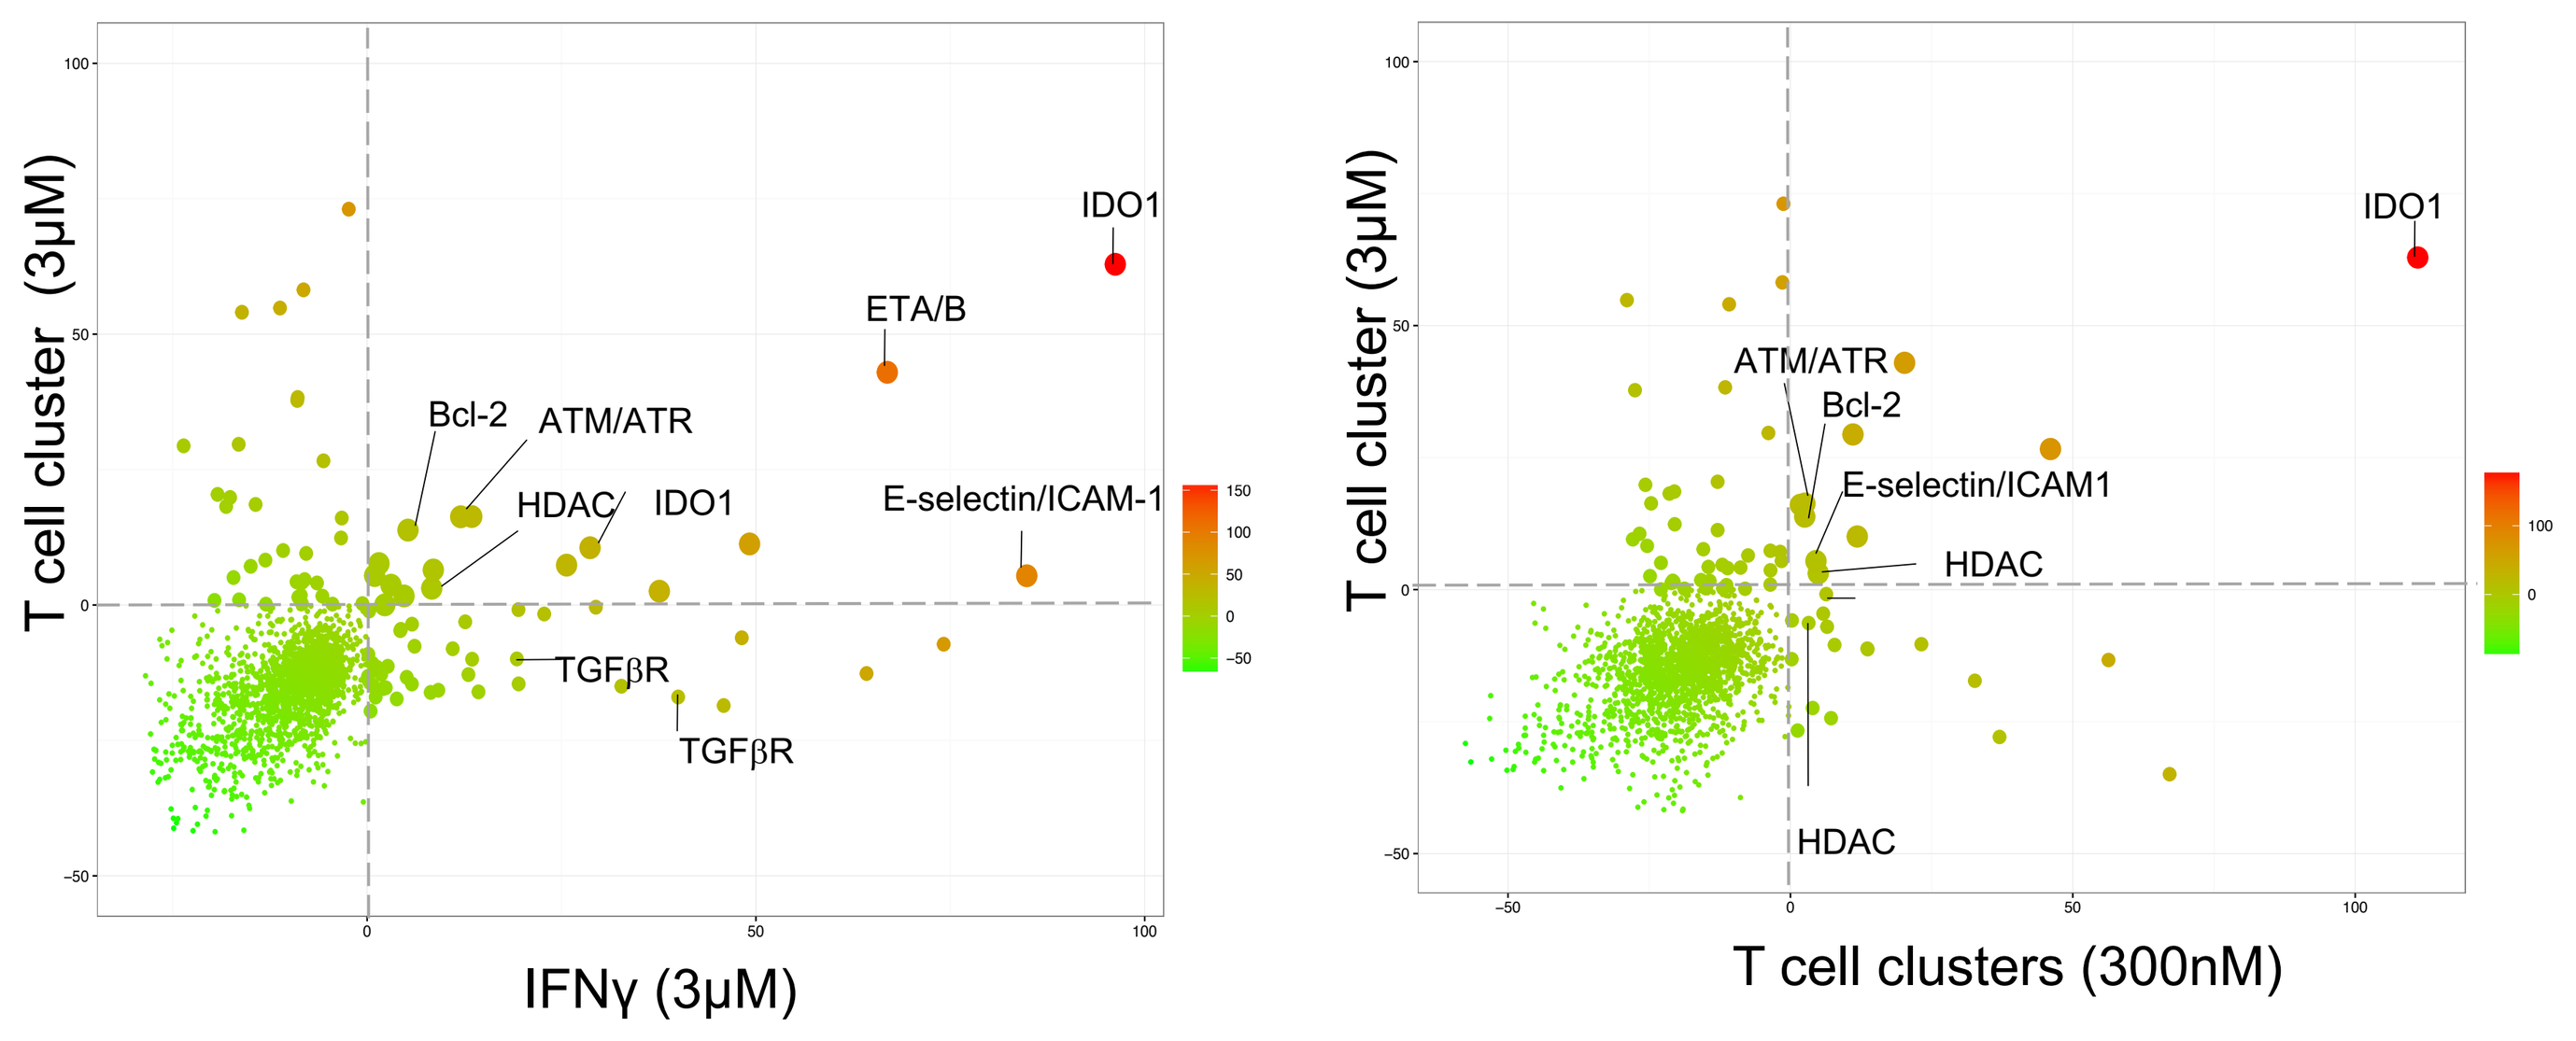
T cell cluster (3μM)
IFNγ (3μM)
IDO1
ETA/B
Bcl-2
ATM/ATR
HDAC
E-selectin/ICAM-1
TGFβR
T cell clusters (300nM)
E-selectin/ICAM1

Compound	Target	Pathway
INCB024360	IDO1	Metabolic
Macitentan	Endothelin Receptor	Growth factor
A-205804	E-selectin/ICAM-1	TNFα/NFκB
ABT-263	Bcl-2	Apoptosis
VE-822	ATM/ATR	DNA damage
AZD2858	GSK3	Metabolic
RepSox	TGFβR	Growth factor
Entinostat	HDAC	Epigenetic

Bubble chart for the hit analysis of the proof of principle run. A compound library containing 1902 different molecules was screened for the ability to revert A549-mediated immunosuppression. As a proof of principle, we screened immunosuppressive A549 cells using two different compound concentrations (300 nM and 3 μM). Cells, were incubated with compounds and CD3+ T cells as described before. At the end of the 5-day workflow, we assessed T cell clusters and INFγ secretion. Bubble charts show the relation between T-cell clusters and IFNγ secretion using 3μM concentration of the compounds (left panel) and between T cell clusters at 3 μM and 300 nM concentration of the compounds (right panel). Red colour and larger bubble size indicate a higher confidence on the hit identification.

SCREEN CONFIRMATION

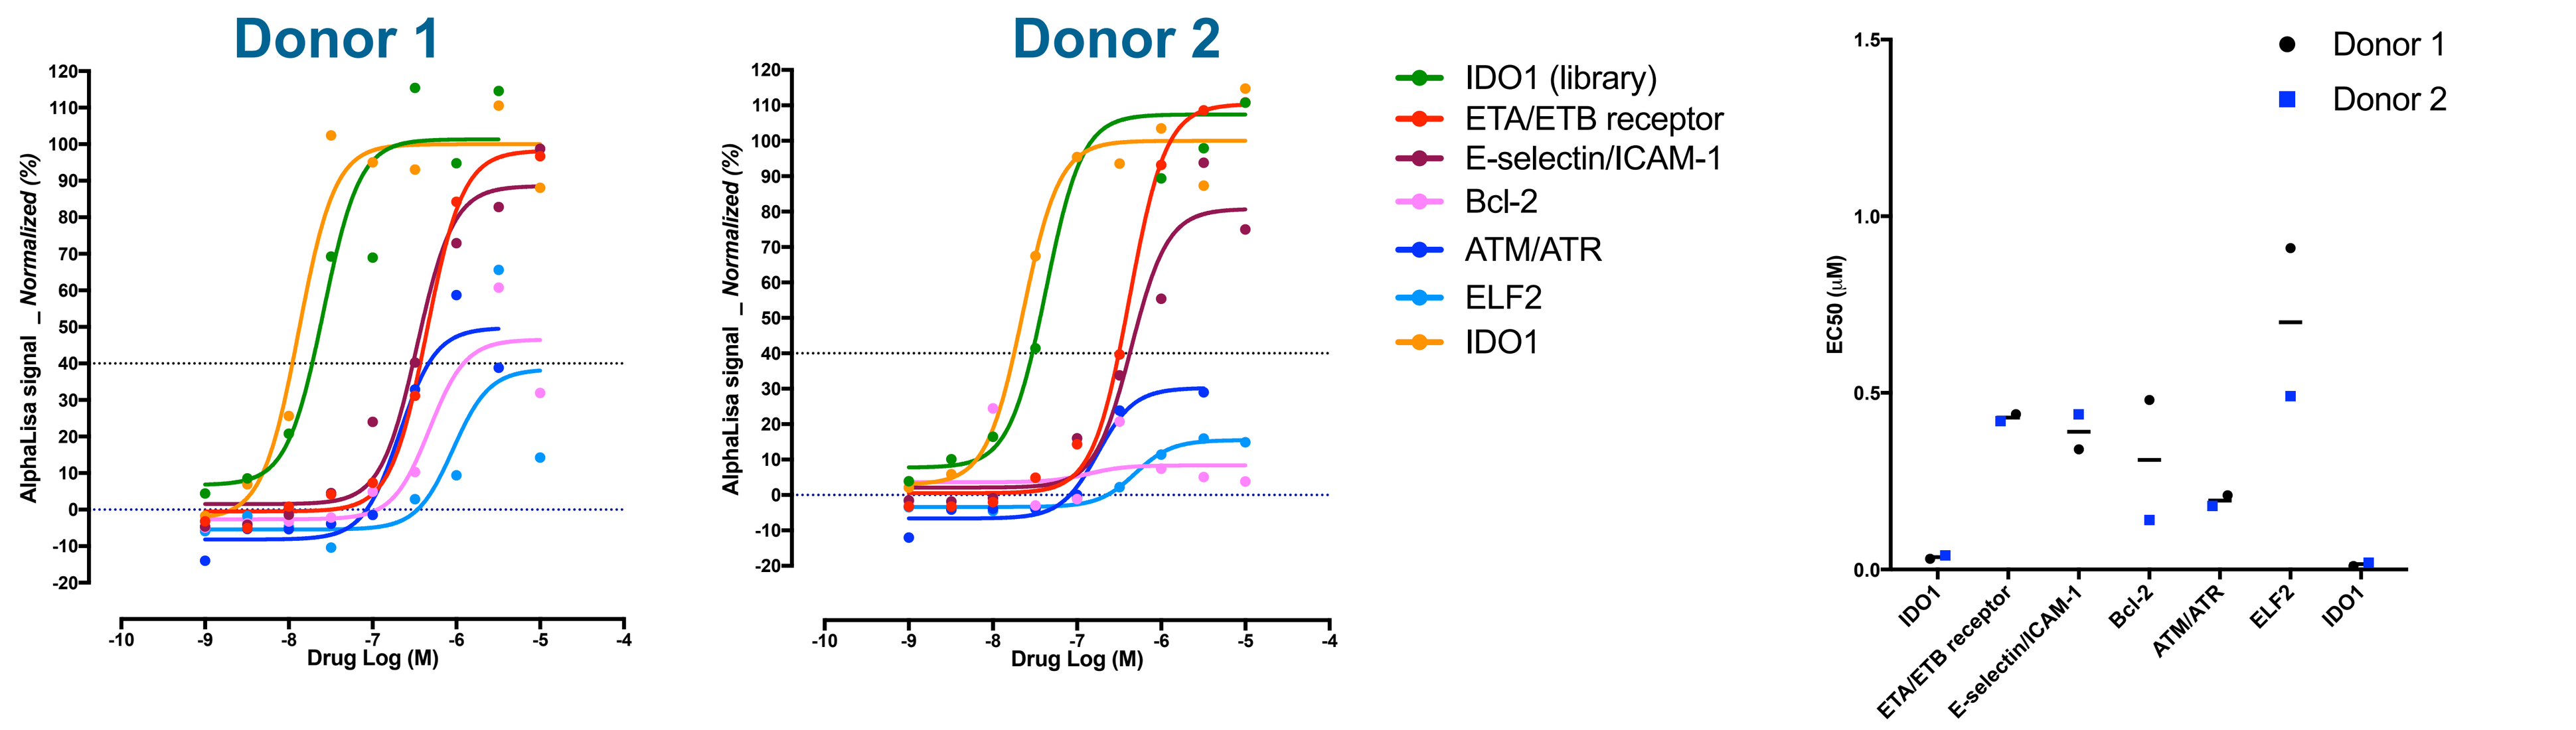
Donor 1
Donor 2
AlphaLisa signal _ Normalized (%)
Drug Log (M)
IDO1 (library)
ETA/ETB receptor
E-selectin/ICAM-1
Bcl-2
ATM/ATR
ELF2
IDO1
EC50 (μM)
Donor 1
Donor 2

Proof of concept confirmation run. Positive compounds resulting from the proof of concept screen were tested for their ability to inhibit A549-mediated immunospression in a dose-dependent manner using CD3+ T cells from two different donors. Shown are the plots of the normalized INFγ secretion (left and middle panels). Robustness of EC_{50} values across CD3+ T cells donors is shown on the right panel.

CONCLUSIONS

- We designed an affordable, automated assay system to identify novel immuno-oncology targets.
- The assay has been validated by performing a medium-throughput screen using immunosuppressive A549 and CD3+ T cells.
- Our screen successfully detected IDO1 inhibitors present in the Selleck library.
- Additionally, we identified hits representing various signaling pathways for their ability to revert immune suppression.

iTeos Therapeutics, Gosselies, Belgium